A Lived Experience Framework to Identify and Address Unmet Needs Across the Clinical Study Lifecycle

Friedhoff Sarah R*, Callison Shannon, MacNally Meghan*, MacQueen Rachael*, Mikell Marisha***, Porco Dania MD*, MacDonald Karla***

*Entrada Therapeutics, Boston, MA, USA
**Current affiliation: Calico Life Sciences, San Francisco, CA, USA
***Current affiliation: Ipsen, Cambridge, MA, USA

Abstract

In 2012, the U.S. Food and Drug Administration (FDA) introduced the concept of patient-focused drug development (PFDD) as a "systematic approach" to "better incorporate the patient voice in drug development and evaluation." Yet, although the FDA released a series of guidance documents on PFDD, industry awareness and standardized application remain inconsistent.

Entrada Therapeutics, Inc., is a clinical-stage biopharmaceutical company whose lead programs are in development for the potential treatment of Duchenne muscular dystrophy (DMD). In late 2023, the company began planning two global Phase 1/2 clinical studies for people living with Duchenne. In order to ensure PFDD, Entrada worked with advocates to build a "lived experience framework": a systematic way to inform strategy and planning for Research and Development (R&D) activities. Five key domains were identified: 1) study design, 2) meaningful outcome measures, 3) recruitment and awareness, 4) participation and retention, and 5) communication and transparency.

From October 2024 to present day (October 2026), Entrada has utilized this framework to gather comprehensive experiential data from a global, independent advisory board of community representatives. This lived experience framework has enabled critical insights, informing inclusion/exclusion criteria, exploratory endpoints, study design, number and timing of assessments, age-stratified retention materials and participant comfort items.

Initial feedback from community members, advocates and study sites confirms perceived value from the decisions influenced by community advisory board (CAB) recommendations, demonstrating how working in partnership with the patient community to integrate lived experience systematically can surface unmet needs and support more patient-centered studies.

Introduction

Beyond growing regulatory precedent and scientific rigor, there is a clear business case for PFDD: data show that patient-centric studies can improve outcomes and save companies time and money:

- It is estimated that each day a company extends the timeline for a clinical study could cost between $600k and $8M in forgone product sales.[3]
- One study found that patient-centric rare disease studies recruited 100 participants in one-fifth the time required by non-patient-centric rare disease studies.[1]
- Avoiding a single substantial amendment in a typical protocol may help prevent three months of additional study time and $141k in median direct costs per amendment in Phase 2 studies.[2]

Looking to the Duchenne space, where Entrada's lead programs are focused, one biotech estimated that the Duchenne community helped prevent 23 amendments to study protocols over their eight years of engagement.[3] With these statistics in mind, this means significant potential time savings (avoiding 5.75 years in protocol amendment delays) and $3.2M potential savings in protocol amendment costs.

In alignment with Entrada's robust commitment to PFDD, the company's Patient Advocacy team spearheaded a cross-functional initiative to build a lived experience framework, from which to gather mixed method data that would both inform strategy and R&D planning activities.

MAT-0066 V1 October 2026

Methods

Constructing a Lived Experience Framework

Entrada conducted qualitative interviews with over 15 advocacy leaders within the U.S. and Europe to understand where community representatives had seen companies succeed or struggle when advancing Duchenne clinical studies. These interviews resulted in the emergence of five key areas, which became Entrada's lived experience framework.

Study design | Outcome measures | Study awareness & recruitment | Study participation & retention | Communication & transparency

Insight Generation

One tenet of Entrada's stakeholder outreach was engaging the World Duchenne Organization's Duchenne CAB. The CAB is an independent, autonomous and international group of trained community representatives from 12 different countries across the world who provide collective knowledge and expertise to industry in order to accelerate research, development, clinical studies and access to effective treatments for DMD worldwide.

CAB Geographic Representation

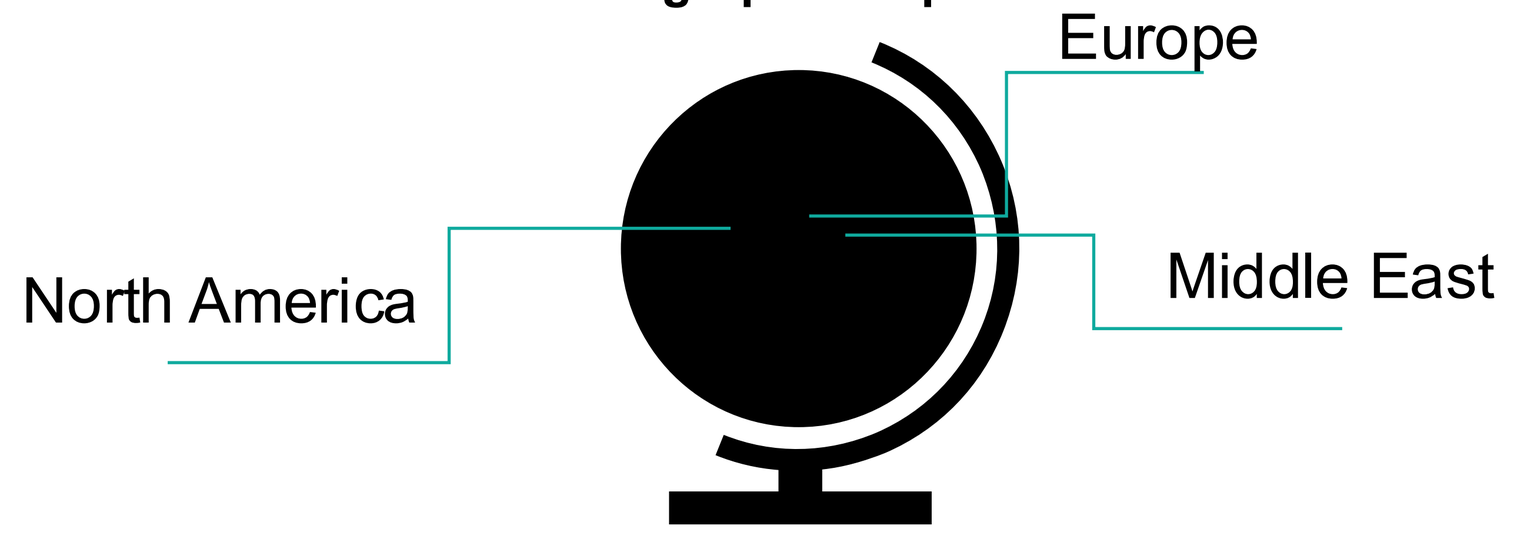
Europe
North America
Middle East

Aligned to the FDA's "Patient-Focused Drug Development: Collecting Comprehensive and Representative Input Guidance," Entrada has met with the CAB five times between 2024 and 2026. The meetings focused on gathering input on study design and outcome measures, participation and recruitment support and expectations for transparent and timely sponsor communication.

CAB meetings were conducted in a focus group format where individual participants answered open-ended questions posed to the group. One meeting required CAB members to read information prior to attending the in-person meeting.

Dimensions of value measured include:

- Participant Experience – Does the change make the study experience better for participants and caregivers?
- Trust and Confidence – Does the change strengthen positive perceptions of the study and sponsor?
- Practical Considerations – Does the change address time, cost, travel or logistical challenges?

Results

Study Design

When evaluating drafts of Entrada's Phase 1/2 protocols, CAB members identified multiple areas where there would be unintended burden to the study participant or their care partner. Recommendations included ways to minimize or eliminate this burden, some of which required external partners to implement.

Value Dimension	Result of Implementing Recommendation	Cross-functional partners
Participant Experience	Reduced number and frequency of questionnaires	Clinical Operations, Health Economics and Outcomes Research
Practical Considerations	Timelines selected to enable access to open label extension (OLE) with minimal-to-no gap	Clinical Operations, Clinical Development
Practical Considerations	Option for centralized travel support (concierge service)	Clinical Operations, Contract Research Organization (CRO)
Participant Experience	Number, location, type and size of muscle biopsies	Clinical Operations, Clinical Development

Outcome Measures

According to the FDA, "A patient reported outcome (PRO) is a measurement based on a report that comes directly from the patient about the status of the patient's health condition without amendment or interpretation." Patient-informed PROs ensure that what is being measured aligns with what is clinically meaningful and humanistically meaningful. CAB members encouraged the use of PROs as appropriate to the stage of development, which give insight into how patients feel and function.

Recommendation	Was Recommendation Implemented?	Outcome
Balance the stage of study with burden of assessments	Yes	Defer PROs to later stages of drug development where population is less variable and tools will be more sensitive to detect meaningful change

Study Awareness and Recruitment

With an inclusion age between ages 4-20 years and a planned global site footprint across the UK and Europe, the CAB emphasized a comprehensive approach to empower and inform participants and care partners of all ages.

Recommendation	Outcome
Study website available in multiple languages	Designed to ensure equitable access to information, the websites enable people from different geographies to make informed decisions about participation, thus reducing common linguistic and information barriers and barriers to enrollment and enabling more diversity in participation.
Adult study overview video	In support of visual learning styles, video explainers were created, in addition more traditionally used print and digital recruitment materials.
Child study overview video	Age-appropriate videos were also created to ensure the participant, regardless of age, felt aware, informed and empowered.

Study Participation and Retention

Feedback from the community focused on creating materials that would be informative, empowering and appropriate for all involved in the study, including young children, adolescents and care partners.

Consent	Participation	Retention
• ICF/assents • Adult study overview video • Child study overview video	• Study sticker chart (4-6 years) • Study sticker visit guide (7-11 years) • Study guide (12+ years) • School notification letter • Welcome cards • Thank-you cards	• Custom backpack with fidget toys, language agnostic games, a blanket and a device holder • Graphic patch for each long-term extension (LTE) visit with a corresponding illustrated postcard, animated video and coloring page

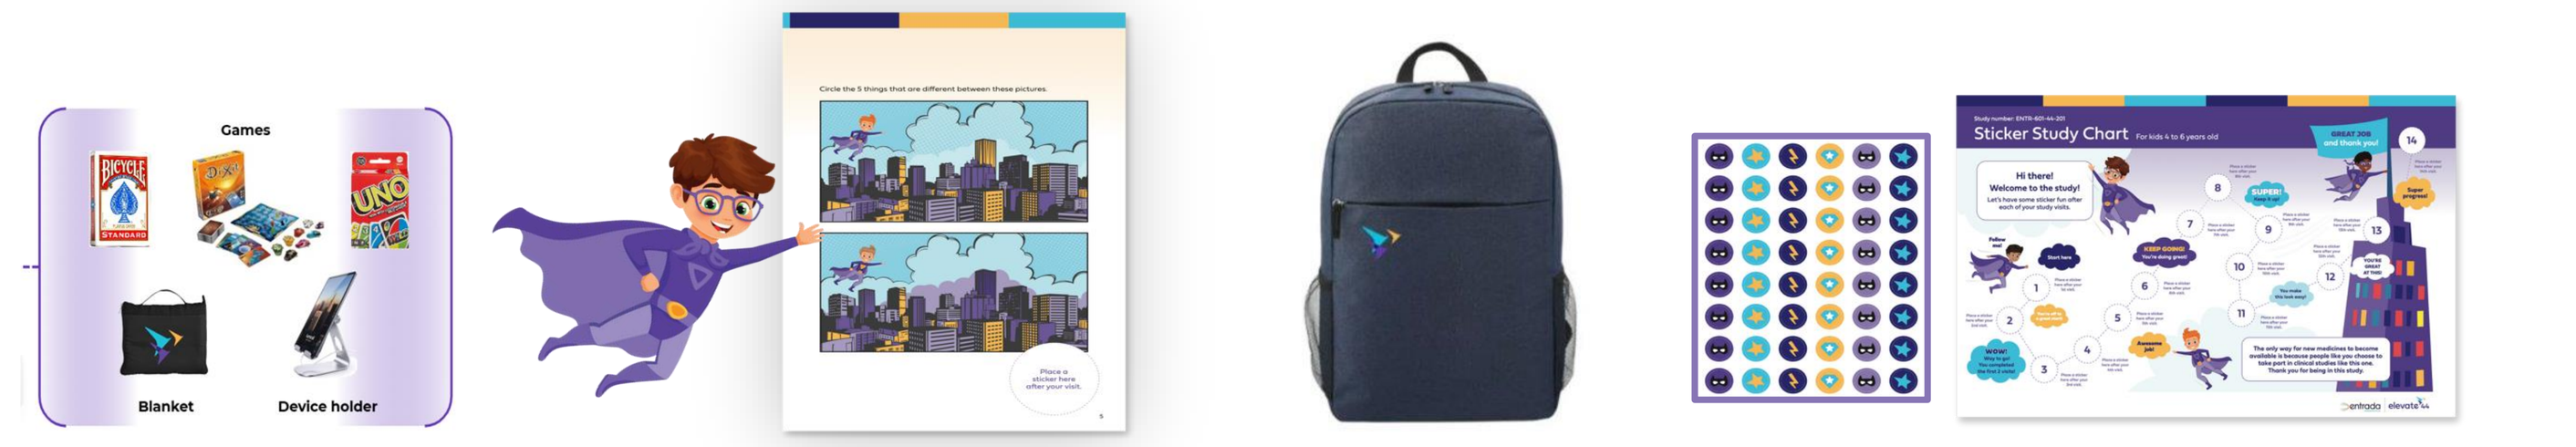
Games
Blanket
Device holder
Sticker Study Chart

"I very much like the letter to school; I have not had one of these supplied for previous studies but is a great idea and very thoughtful of the Sponsor!" — Email from Research Nurse

Communication and Transparency

Recognizing that clear, timely and transparent communication is essential to building trust and confidence, Entrada worked closely with patients, care partners and advocates to establish a systematic approach to sharing updates on the progress of its Duchenne programs.

Community Updates

All program-related company news releases have a simultaneously published plain-language "Community Update," which is sent to advocacy groups and individuals who opt in.

Quarterly Newsletter

To keep the community engaged and informed, Patient Advocacy provides a quarterly newsletter which includes updates on studies, highlights from recent events and information on other news and initiatives.

Entrada integrated an automated translation feature on the web versions of all communications, enabling readers to view content in 50+ languages.

Conclusions

Implementation of a structured lived experience framework enabled the systematic integration of patient and care partner perspectives into clinical study planning and execution. Through ongoing collaboration with the Duchenne community, insights were translated into actionable changes across study design, outcome measures, recruitment, participation, retention and communications.
Early qualitative and quantitative feedback from CAB members, study sites and community members suggests these efforts are positively perceived. Ongoing evaluation will assess the perceived value and impact of these changes, helping to build an evidence base for the role of patient-focused drug development in rare disease research.

References

[1]The Economist Intelligence Unit. The Innovation Imperative: The Future of Drug Development. Part I. 2018.
[2]Getz KA, et al. Ther Innov Regul Sci. 2016;50(4):436-441.
[3] Levine DS, Beek G, Bichell TJ, et al. Early and Often: Reimagining Patient Community Engagement to Improve Clinical Trials Feasibility. Global Genes Corporate Alliance; 2025.

Acknowledgments

Entrada would like to thank each of the Duchenne CAB members, CAB chair, CAB coordinator, community advisors and patient advocacy organizations for their insights, feedback and recommendations.

Disclosure

Entrada Therapeutics has provided grant funding and/or sponsorship support to WDO. WDO is an independent organization and retains sole control over its programs, activities, and positions.

NORD Breakthrough Summit 2026
Contact: patientadvocacy@entradatx.com
Entrada Therapeutics, Inc., Boston, MA