4.50eP

Pharmacokinetic and pharmacodynamic translation of ENTR-601-44, an investigational therapy for patients with exon 44 skipping-amenable DMD

Ridhi Parasrampuria[1], Eric T. Williams[1], Wenlong Lian[1], Nick Long[1], Kalina Dimova[1], Nanjun Liu[1], Dania M.D. Porco[1], Divya B. Reddy[1], Terrance A. Stadheim[1], Natarajan Sethuraman[1], Kiran Patki[1]

[1]Entrada Therapeutics, Boston, MA, USA

Background

- Currently approved antisense phosphorodiamidate morpholino oligomer (PMO)–mediated exon-skipping therapies for Duchenne muscular dystrophy (DMD) face challenges related to limited intracellular delivery[1,2]
- The Endosomal Escape Vehicle (EEV™) Platform was designed to improve cellular uptake and endosomal escape of therapeutic modalities, including PMO delivery to muscle[1,3,4]
- ENTR-601-44 is an EEV-PMO conjugate intended to target dystrophin pre-mRNA in the nucleus and induce exon 44 skipping, with the goal of restoring expression of near full-length and functional dystrophin protein
 - In nonclinical studies, ENTR-601-44 induced exon 44 skipping, increased dystrophin production in patient-derived cells and in a DMD mouse model bearing an exon 44 skipping-amenable dystrophin mutation, and demonstrated exon 44 skipping in nonhuman primates (NHPs)[5,6]
 - In clinical studies of ENTR-601-44 in adult healthy volunteers (HVs) and in pediatric participants with DMD, ENTR-601-44 has had a generally favorable safety profile, with no serious adverse events and no clinically relevant changes in renal function or kidney injury biomarkers observed to date[7,8]
- Here, we present pharmacokinetic (PK) and pharmacodynamic (PD) data for ENTR-601-44 from NHPs and participants enrolled in clinical studies of ENTR-601-44 (HVs and pediatric participants with DMD)

Objective

- To evaluate the translational PK/PD profile of ENTR-601-44 based on data from NHPs and early clinical development in HVs and pediatric individuals with exon 44 skipping-amenable DMD

Methods

- Properties of ENTR-601-44 and its final PMO-containing metabolite were characterized in several studies, as follows:
 - Four NHP studies in either adult (34 to 44 months of age at study initiation) or juvenile (~14 months of age at study initiation) animals
 - A single-dose study in adult animals (intravenous [IV] bolus)
 - Two repeat-dose studies in adult animals (five dose levels administered either every six weeks [Q6W] or every eight weeks [Q8W] by IV infusion)
 - A repeat-dose study in juvenile animals (three dose levels; Q6W IV infusion)
 - The Phase 1 ENTR-601-44-101 study in adult HVs (0.75−6 mg/kg IV infusion, single dose)[7]
 - The ongoing, double-blind, multiple-ascending dose (MAD) period of the Phase 1/2b ELEVATE-44-201 study in participants with exon 44 skipping-amenable DMD (Cohort 1: ENTR-601-44 6 mg/kg or placebo IV infusion, administered Q6W for three doses)[8]
- Assessments included longitudinal plasma PK and 0 to 24-hour urine PK collected on multiple days post-dose; skeletal muscle biopsies were collected to evaluate both PK and PD assessments (eg, exon 44 skipping)
 - Validated liquid chromatography with tandem mass spectrometry (LC-MS/MS) was used to measure the concentrations of ENTR-601-44 and its final PMO-containing metabolite in plasma, urine, and muscle biopsies
 - Total urinary excretion over multi-week periods was determined by calculating the mass of the final PMO-containing metabolite in each discrete 24-hour pool and chronologically summing them
- Data were summarized descriptively

Conclusions

- The EEV-PMO ENTR-601-44, which is under investigation for the potential treatment of individuals with DMD amenable to exon 44 skipping, demonstrated dose-dependent PK
- Supra-proportional uptake of the final PMO-containing metabolite in muscle and exponential urinary elimination in NHPs and HVs were also observed
- Results suggested differences between HVs and NHPs in plasma PK (two- to three-fold higher in HVs) and muscle PK (~ three-fold higher in HVs) at matched doses
- The final PMO-containing metabolite of ENTR-601-44 demonstrated a long residence time in muscle of pediatric participants with DMD amenable to exon 44 skipping
- Juvenile vs adult PK differences appeared to be comparable in NHPs and humans
- In juvenile NHPs, exon skipping increased in a nonlinear manner, with a pronounced effect at the highest dose, which may result in greater exon skipping and other PD effects with ENTR-601-44 doses above 6 mg/kg in participants with DMD in the ongoing ELEVATE-44-201 study

Results

Dose-dependent plasma PK translates from nonclinical study to clinical healthy volunteer study

- In both NHPs and HVs, ENTR-601-44 had a dose-dependent plasma exposure (**Figures 1A** and **1B**)[7] and rapid systemic clearance
 - The plasma exposure of ENTR-601-44 in adult HVs appeared to be approximately two- to three-fold higher vs levels in adult NHPs with body weight-matched doses
- No accumulation in plasma on repeated dosing was evident in participants with DMD (data not shown)
- The PMO-containing metabolite persisted in plasma longer vs ENTR-601-44 in NHPs (data not shown), HVs (**Figure 1C**; half-life of ENTR-601-44, 2−4 hours; half-life of the final PMO-containing metabolite, >100 hours), and participants with DMD (data not shown)

Figure 1. Plasma PK of ENTR-601-44 and final PMO-containing metabolite in NHPs and HVs

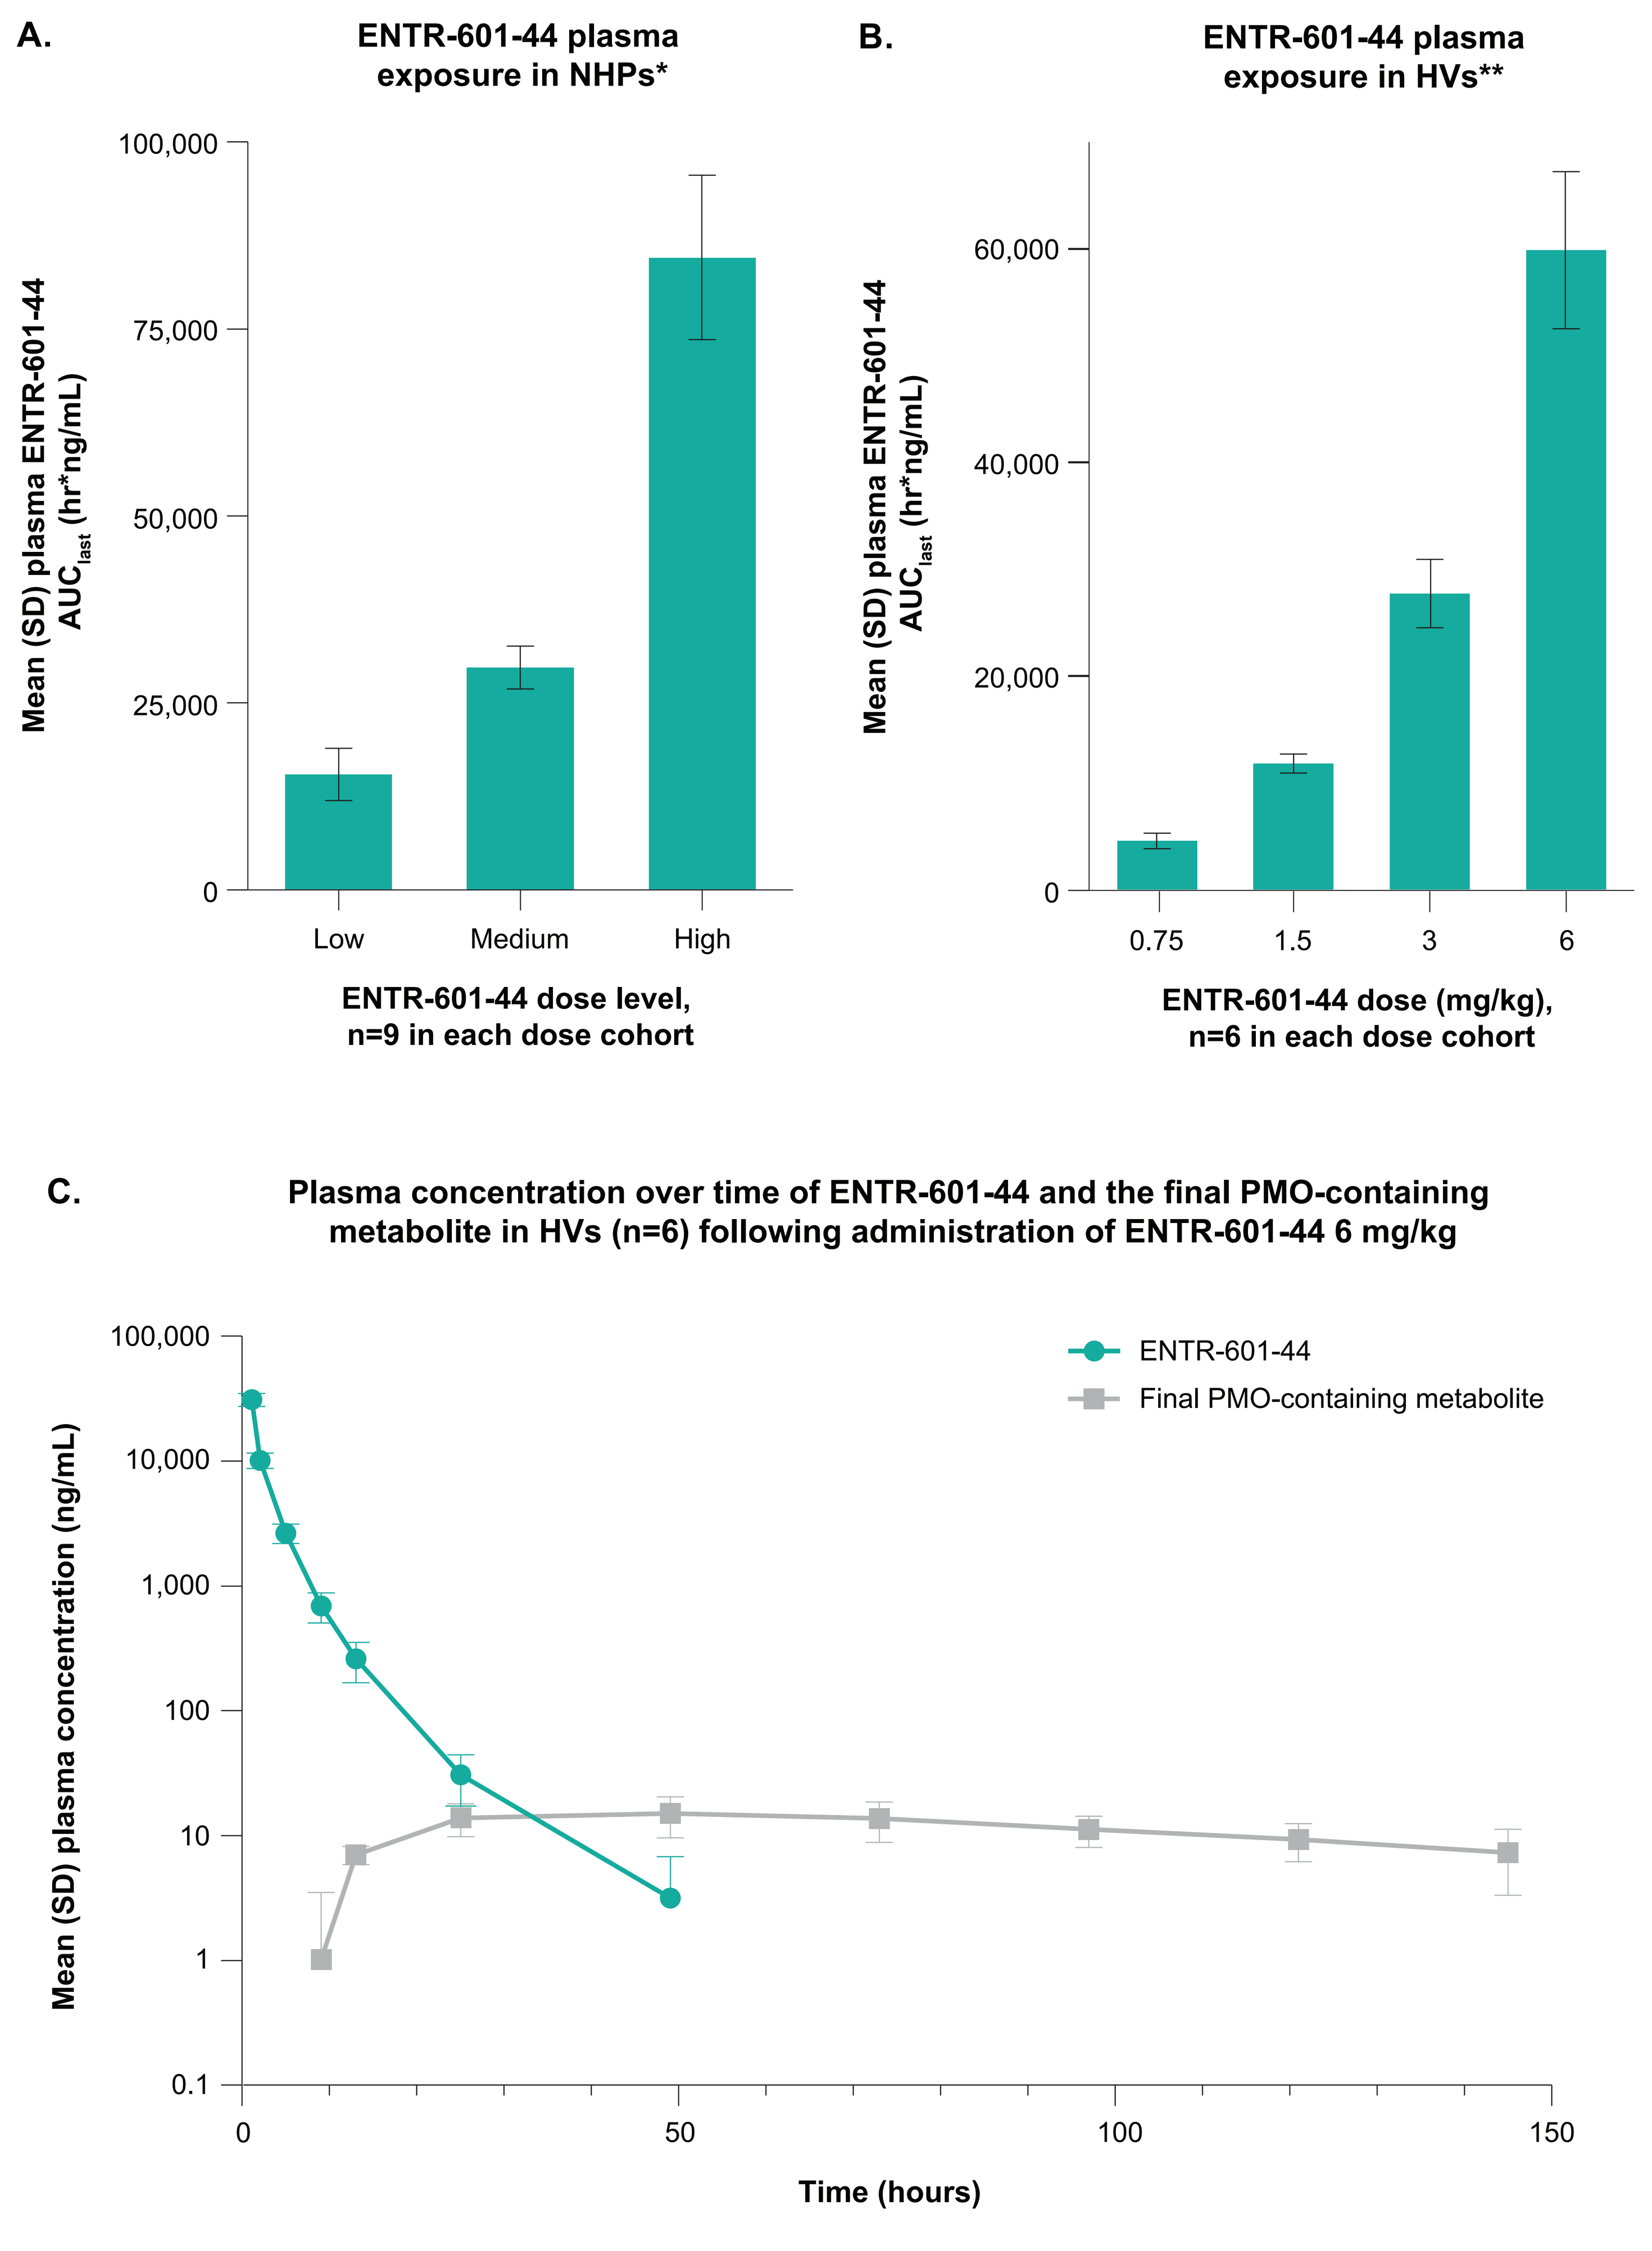
A.
ENTR-601-44 plasma exposure in NHPs*
Mean (SD) plasma ENTR-601-44 AUC$_{last}$ (hr*ng/mL)
100,000
75,000
50,000
25,000
0
Low
Medium
High
ENTR-601-44 dose level, n=9 in each dose cohort
B.
ENTR-601-44 plasma exposure in HVs**
Mean (SD) plasma ENTR-601-44 AUC$_{last}$ (hr*ng/mL)
60,000
40,000
20,000
0
0.75
1.5
3
6
ENTR-601-44 dose (mg/kg), n=6 in each dose cohort
C.
Plasma concentration over time of ENTR-601-44 and the final PMO-containing metabolite in HVs (n=6) following administration of ENTR-601-44 6 mg/kg
Mean (SD) plasma concentration (ng/mL)
100,000
10,000
1,000
100
10
1
0.1
0
50
100
150
Time (hours)
ENTR-601-44
Final PMO-containing metabolite

*ENTR-601-44 administered as a repeat dose Q6W; PK data post first-dose shown; ** Day 1 PK data following a single dose of ENTR-601-44; **AUC**: Area under the concentration-time curve; **HV**: Healthy volunteer; **NHP**: Nonhuman primate; **PK**: Pharmacokinetics; **PMO**: Phosphorodiamidate morpholino oligomer; **Q6W**: Every six weeks.

Juvenile vs adult PK differences

- Lower peak concentration (C_{max}; mean 43% lower) and area under the concentration-time curve (AUC; mean 58% lower) were observed in pediatric participants with exon 44 skipping-amenable DMD vs adult HVs (**Figure 2**)
 - Similarly, in NHPs, lower C_{max} (mean ~50% lower) and AUC (mean ~50% lower) were observed in juvenile vs adult animals
- Juvenile vs adult PK differences were generally similar in NHPs and humans (data not shown)

Figure 2. Plasma PK parameters in adult HVs and pediatric participants with DMD following ENTR-601-44 6 mg/kg administration

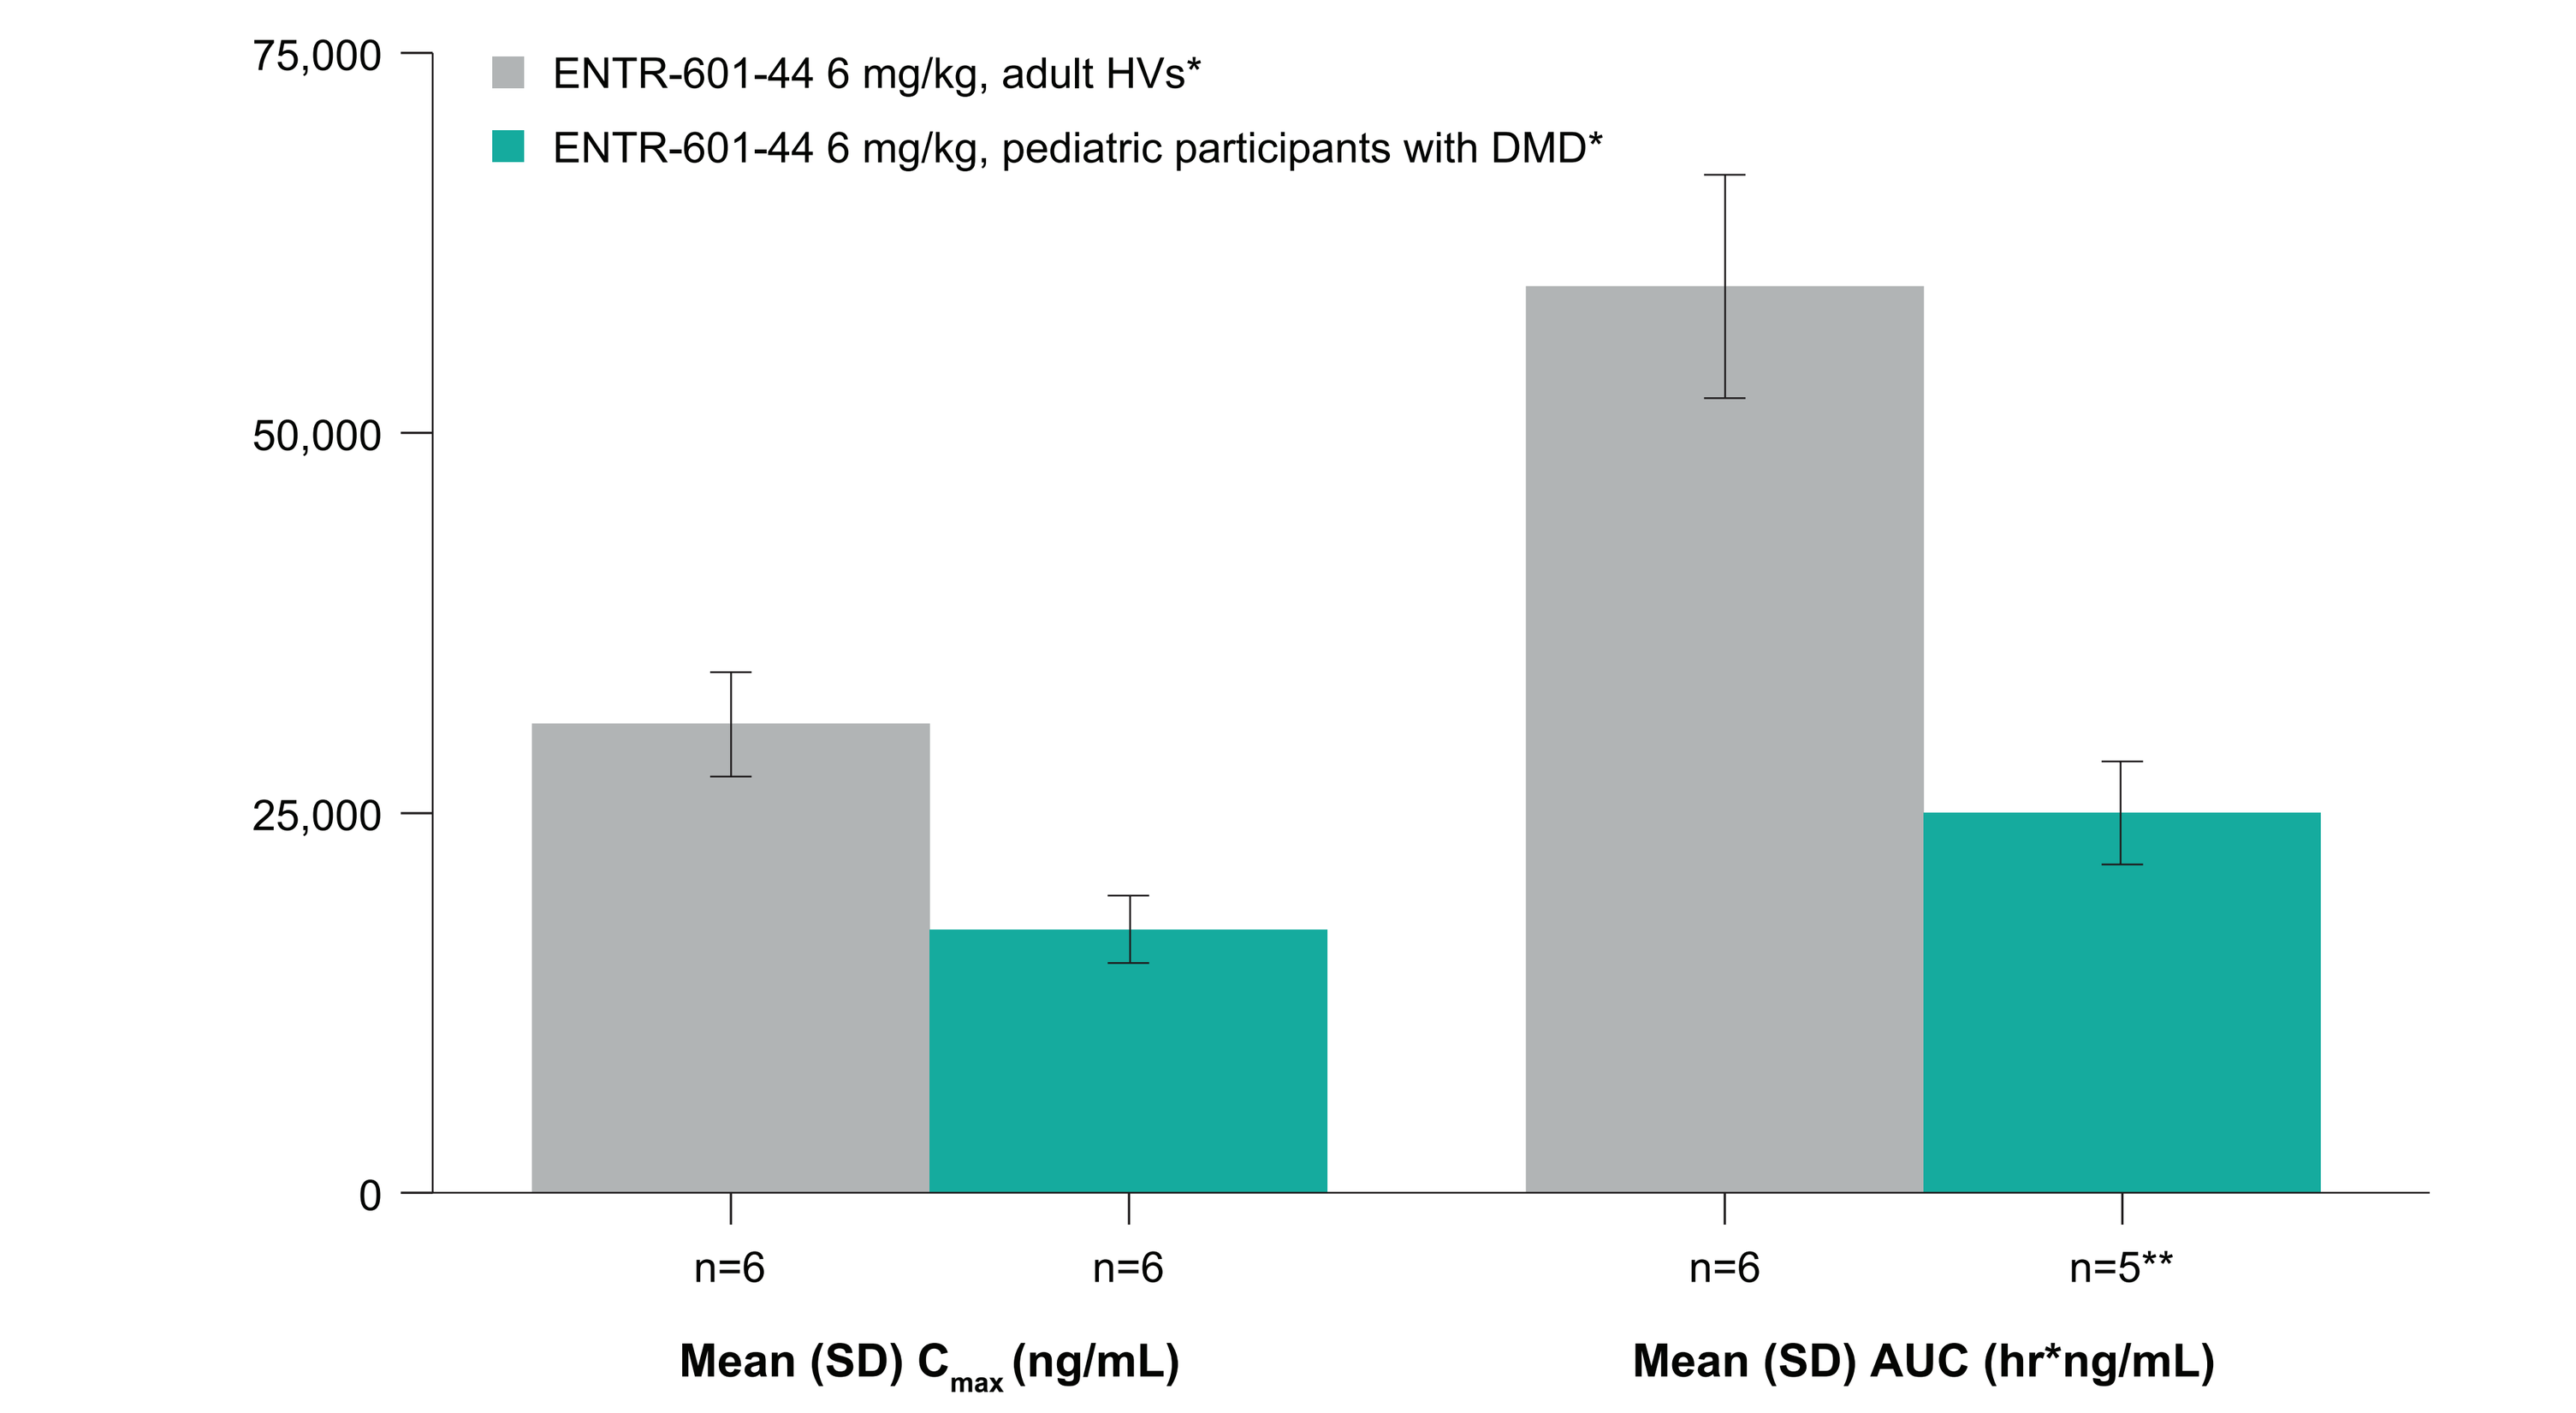
75,000
50,000
25,000
0
ENTR-601-44 6 mg/kg, adult HVs*
ENTR-601-44 6 mg/kg, pediatric participants with DMD*
n=6
n=6
n=6
n=5**
Mean (SD) C_{max} (ng/mL)
Mean (SD) AUC (hr*ng/mL)

*Day 1 PK profile following 6 mg/kg dose of ENTR-601-44; **AUC was not calculable for 1 participant based on an incomplete plasma concentration-time profile due to limited sampling for low body weight; C_{max} is reported for all evaluable subjects; **AUC**: Area under the concentration-time curve; **C_{max}**: Peak concentration; **DMD**: Duchenne muscular dystrophy; **hr**: Hour; **HV**: Healthy volunteer; **PK**: Pharmacokinetic; **Q6W**: Every six weeks.

Dose-dependent delivery of final PMO-containing metabolite to muscle and excretion in the urine in nonhuman primates and healthy volunteers

- Dose-dependent skeletal muscle concentrations of the final PMO-containing metabolite of ENTR-601-44 were observed in both adult HVs and NHPs (**Figure 3**)
 - Mean concentration of the final PMO-containing metabolite in muscle following a single dose of ENTR-601-44 suggested approximately three-fold higher exposure in HVs than in NHPs
- In participants with DMD, concentrations of the final PMO-containing metabolite in muscle biopsy were detectable six weeks after the third dose (data not shown)

Figure 3. Skeletal muscle concentration of the final PMO-containing metabolite after ENTR-601-44 administration

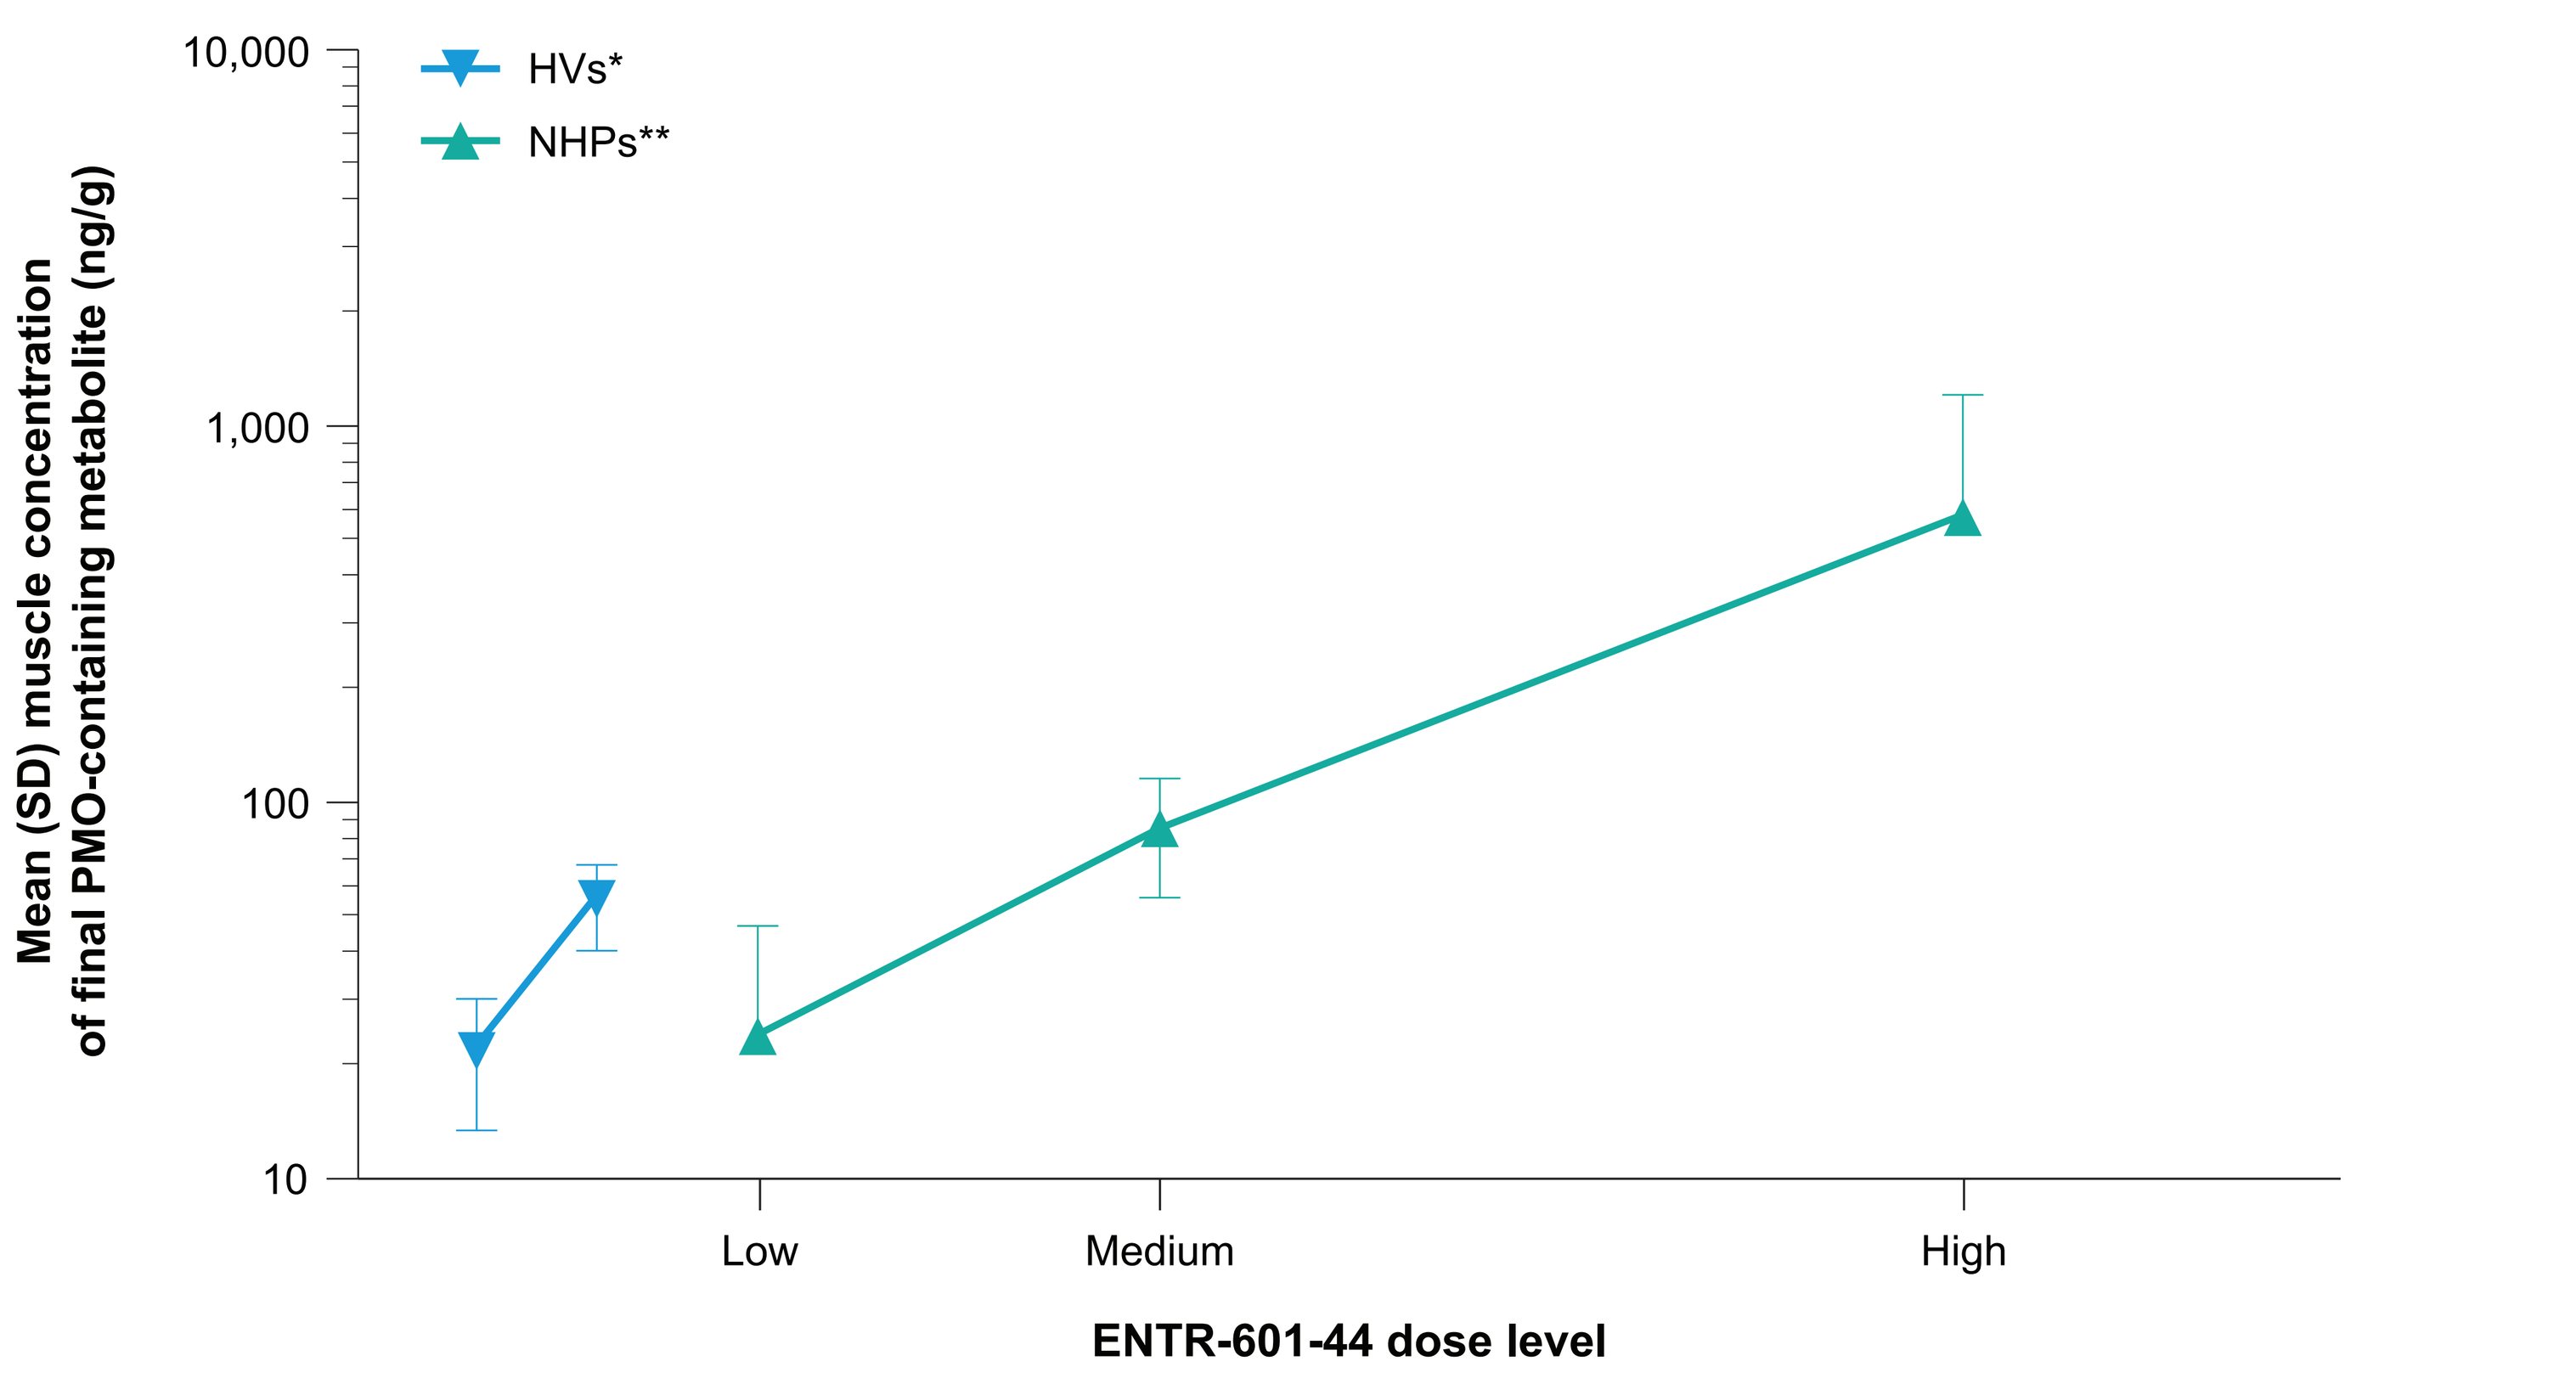
Mean (SD) muscle concentration of final PMO-containing metabolite (ng/g)
10,000
1,000
100
10
HVs*
NHPs**
Low
Medium
High
ENTR-601-44 dose level

*In HVs, data are shown from needle muscle biopsies taken at 72 (±4) hours after a single ENTR-601-44 dose (3 mg/kg, n=3; 6 mg/kg, n=6); **In NHPs, data are shown from a muscle biopsy taken at 48 hours after the first dose (n=3 per group) of a repeat-dose study; **HV**: Healthy volunteer; **NHP**: Nonhuman primate.

- In both NHPs and HVs, the final PMO-containing metabolite of ENTR-601-44 was detected in urine (range of geometric means, 22.9%−50.8%) (**Figure 4**)
- In HVs, for every doubling of dose, there was a more than doubling of metabolite excretion, suggesting saturation of uptake of PMO in the kidney[7]
- Greater urinary excretion of the final PMO-containing metabolite (geometric mean: 50.8%) was observed in HVs at 6 mg/kg than in NHPs (22.9%−24.9%)
- No clinically significant findings related to renal function were noted at 6 mg/kg in HVs[7] or study participants with DMD[8]

Figure 4. Urinary excretion of the final PMO-containing metabolite in HVs and NHPs following single-dose administration of ENTR-601-44

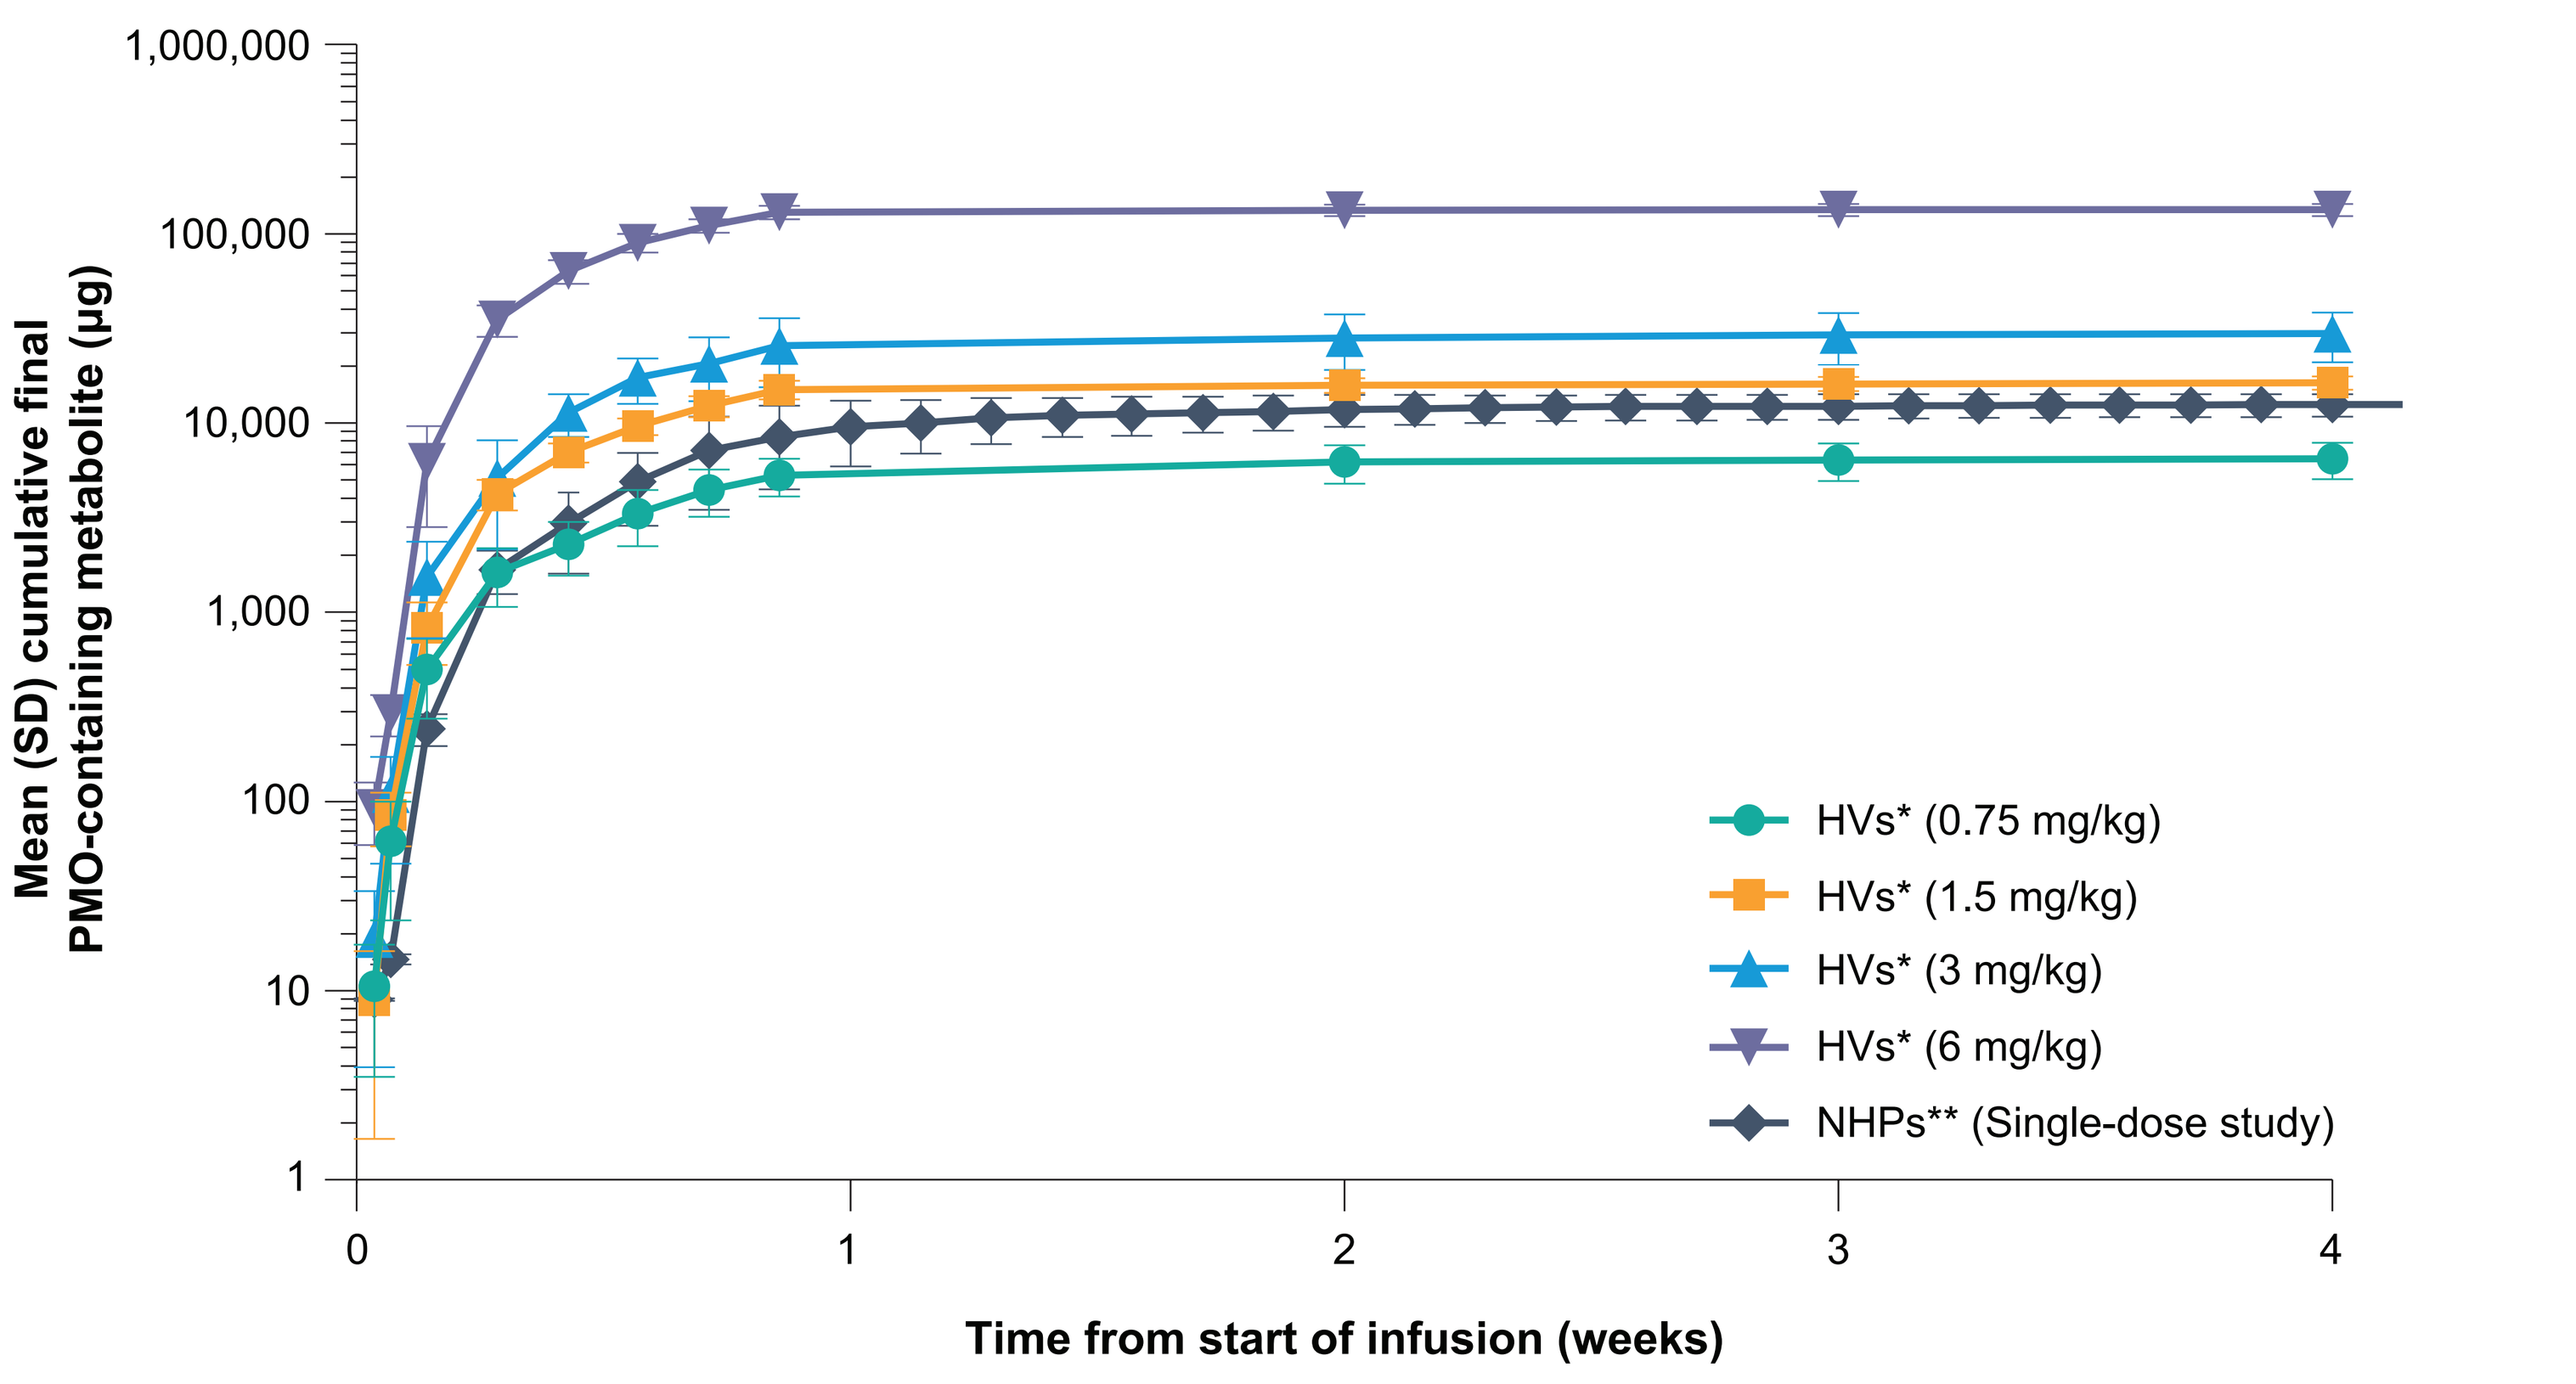
Mean (SD) cumulative final PMO-containing metabolite (µg)
1,000,000
100,000
10,000
1,000
100
10
1
0
1
2
3
4
Time from start of infusion (weeks)
HVs* (0.75 mg/kg)
HVs* (1.5 mg/kg)
HVs* (3 mg/kg)
HVs* (6 mg/kg)
NHPs** (Single-dose study)

*Data from n=6 adult HVs per cohort; **Data from n=2 adult animals; **HV**: Healthy volunteer; **NHP**: Nonhuman primate; **PMO**: Phosphorodiamidate morpholino oligomer.

ENTR-601-44 PK/PD relationship in juveniles

- Dose-dependent exon skipping was observed in juvenile NHPs, with a nonlinear increase evident at the highest dose (**Figure 5**)
- Based on these findings, the higher exposures anticipated in pediatric participants from Cohorts 2 and 3 of the ELEVATE-44-201 study (with ENTR-601-44 doses of ≥12 mg/kg) relative to Cohort 1 may result in greater PD effects

Figure 5. ENTR-601-44 plasma exposure and percentage of exon skipping after ENTR-601-44 administration in juvenile NHPs*

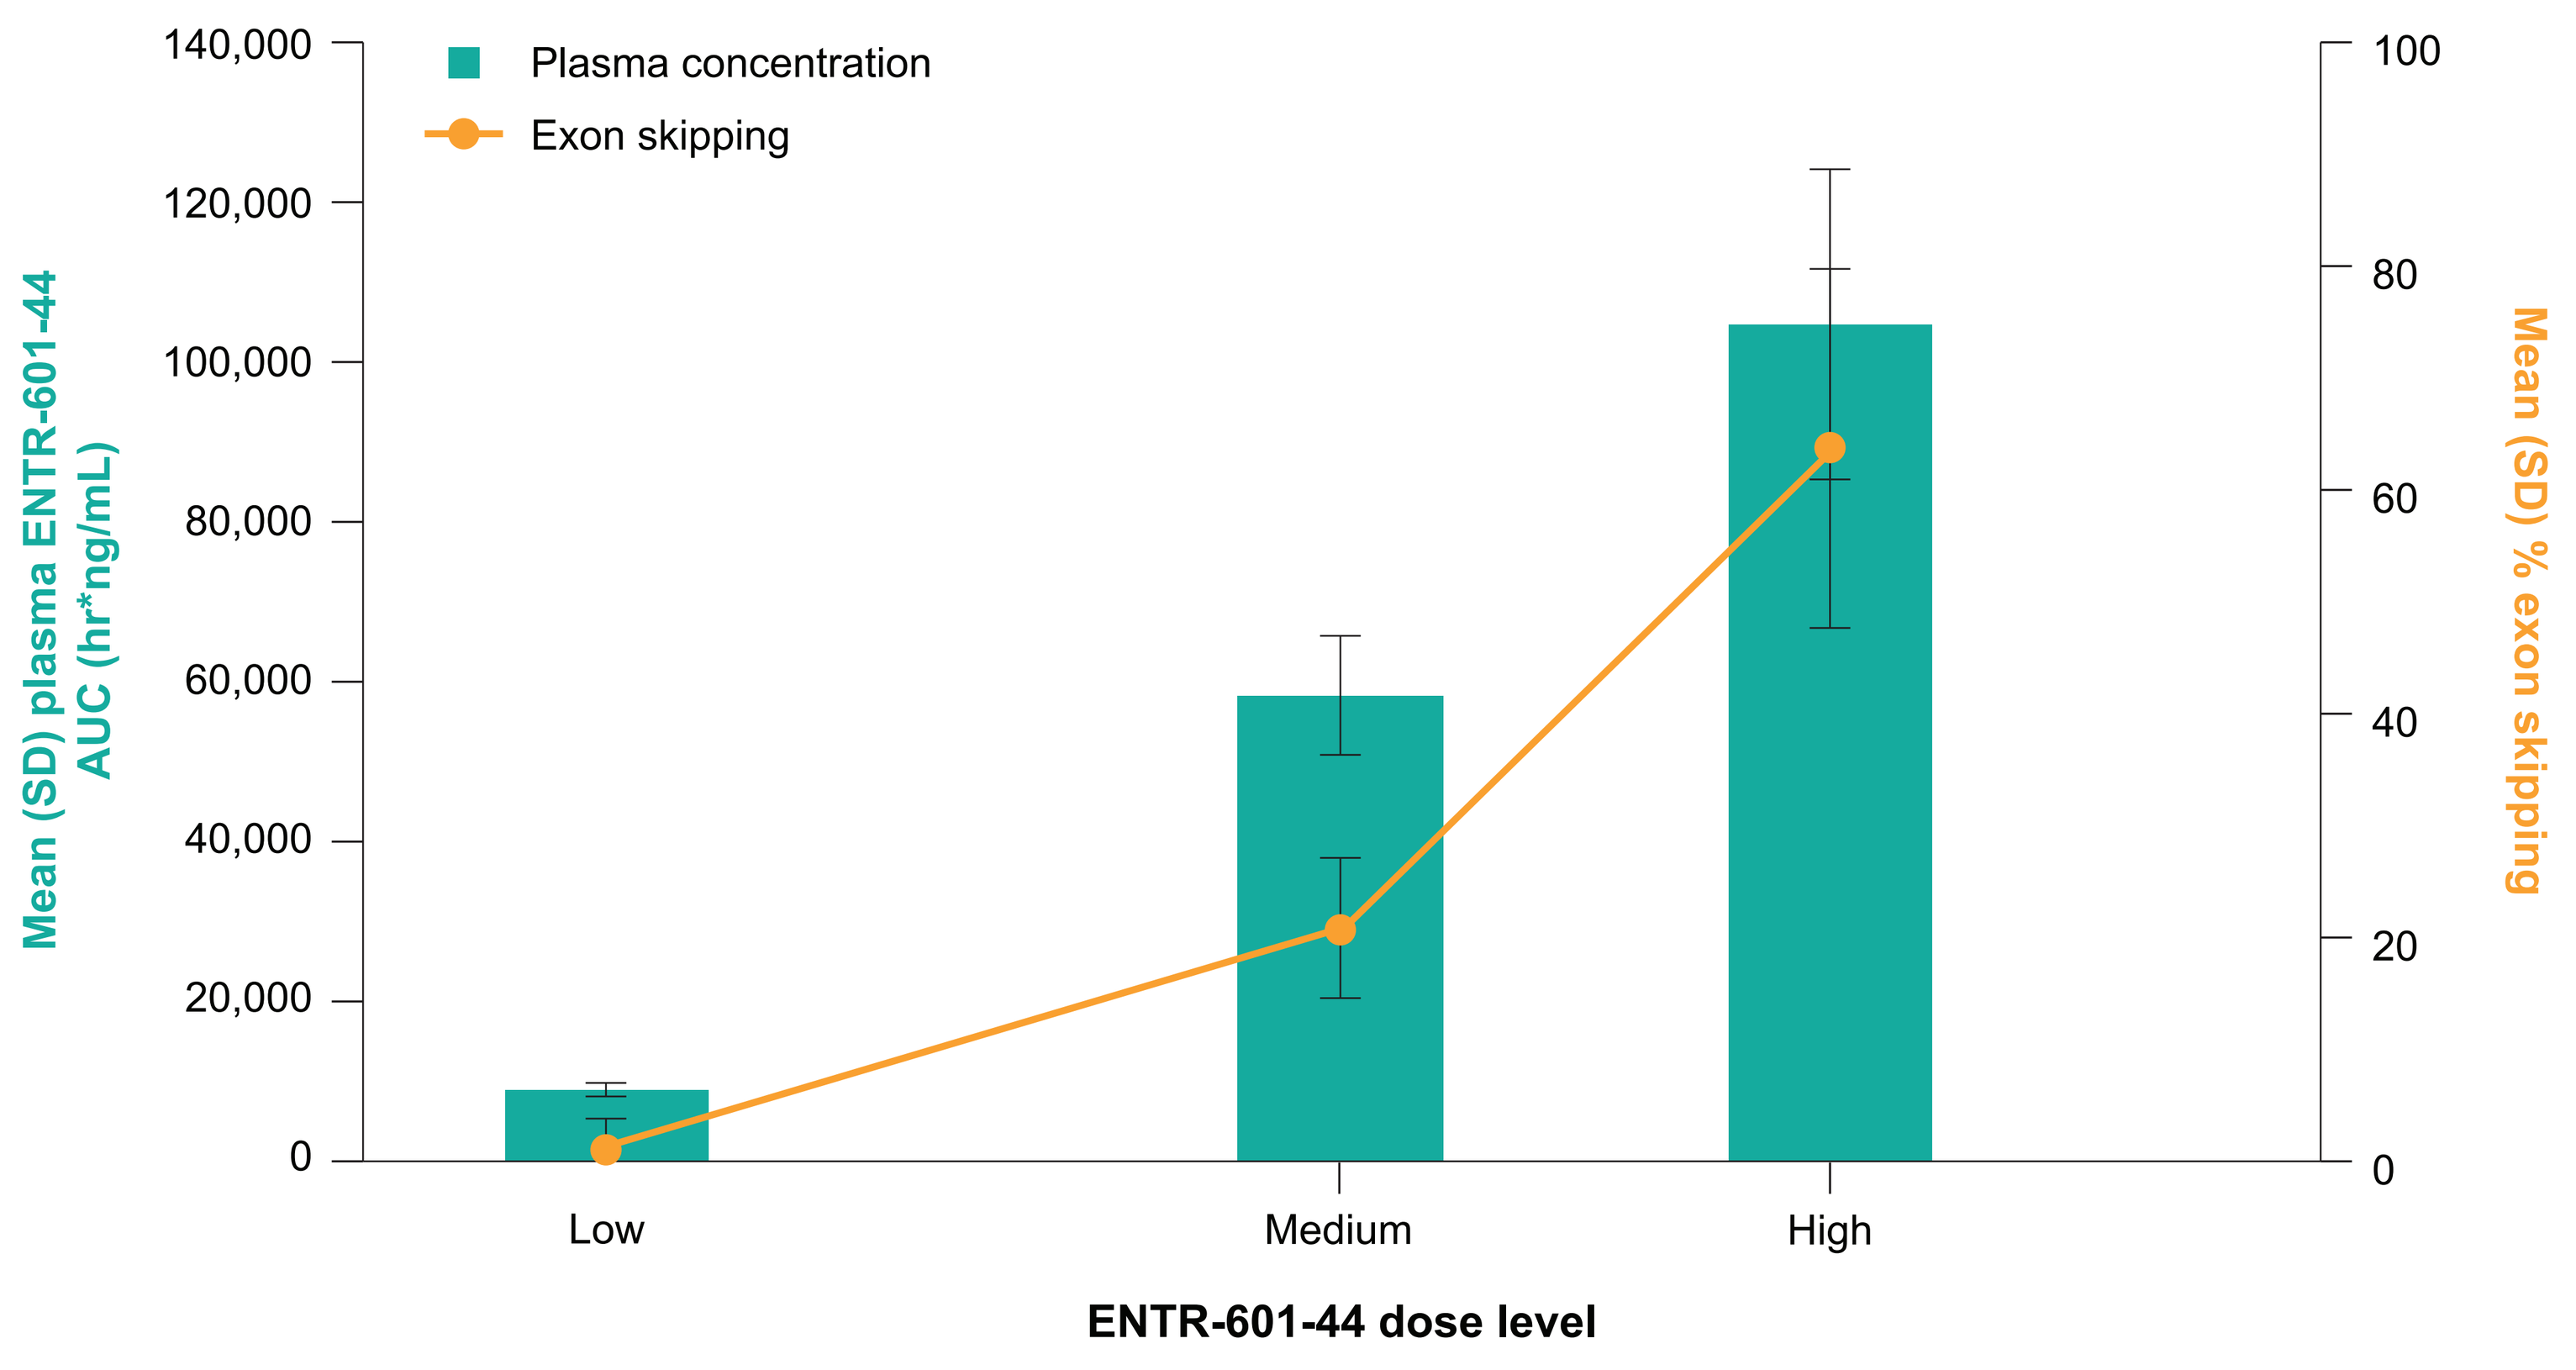
Mean (SD) plasma ENTR-601-44 AUC (hr*ng/mL)
140,000
120,000
100,000
80,000
60,000
40,000
20,000
0
Plasma concentration
Exon skipping
Mean (SD) % exon skipping
100
80
60
40
20
0
Low
Medium
High
ENTR-601-44 dose level

*ENTR-601-44 administered as a repeat dose Q6W (data shown from end of study; n=6 per group; exon skipping was assessed using a muscle biopsy taken from biceps); **AUC**: Area under the concentration-time curve; **NHP**: Nonhuman primate; **Q6W**: Every six weeks.

References

1. Li X, et al. *Mol Ther Nucleic Acids.* 2023;33:273-85. 2. Roberts TC, et al. *Nat Rev Drug Discov.* 2020;19:673-94. 3. Qian Z, et al. *ACS Chem Biol.* 2013;8:423-31. 4. Pei D. *Acc Chem Res.* 2022;55:309-18. 5. Girgenrath M. Presented at: Muscular Dystrophy Association (MDA) Clinical & Scientific Conference; March 13−16, 2022; Nashville, TN, USA. 6. Girgenrath M. Presented at: American Society of Gene & Cell Therapy; May 13−17, 2025; New Orleans, LA. 7. Oldham M, et al. Presented at: 29th Annual Congress of the World Muscle Society (WMS 2024); October 8−12, 2024; Prague, Czechia. 8. Servais L, et al. Presented at: International Congress on Neuromuscular Diseases; July 5−9, 2026; Florence, Italy.

Author Disclosures

RP, ETW, WL, NLo, KD, NLi, DMDP, DBR, TAS, NS, and **KP:** Employees of Entrada Therapeutics, Inc.

Acknowledgments

We thank all the study participants, their families, and the investigators and their study teams. This study is sponsored by Entrada Therapeutics, Inc. (Boston, MA). Medical writing and editorial assistance were provided by Gina Daniel, PhD, of Peloton Advantage, LLC, an OPEN Health company, and funded by Entrada Therapeutics, Inc.

Scan for the poster

31st Annual Congress of the World Muscle Society (WMS 2026); 29 September−3 October 2026; Hiroshima, Japan Contact: medical@entradatx.com Entrada Therapeutics, Inc., Boston, MA | MAT-0040, v1.0, September 2026