Poster #1633

Endosomal Escape Vehicle–Oligonucleotide Conjugates for the Targeted Upregulation and Downregulation of Gene Expression

Mahboubeh Kheirabadi, Xiang Li, Haoming Liu, Patrick G. Dougherty, Matthew Streeter, Amy N. Hicks, Mark Wysk, Ajay Kumar, Jia Li, Anushree Pathak, Kimberli J. Kamer, Nelsa L. Estrella, Arianna Bonilla, Emmanuelle Sizensky, Wenlong Lian, Nanjun Liu, Sara L. Blake, Christopher M. Brennan, Ning Li, Mohanraj Dhanabal, Mahasweta Girgenrath, Ziqing Leo Qian

Entrada Therapeutics, Boston, MA

INTRODUCTION

- Intracellular delivery of oligonucleotides is challenging because of poor cell entry and limited escape from the endosome in the target cell.[1,2]
- To improve cellular uptake and enhance endosomal escape of oligonucleotides and other biologics, we developed the endosomal escape vehicle (EEV)™ delivery platform based on cyclic cell-penetrating peptides (**Figure 1**).
- Here, we assessed the broad applicability of the EEV platform to deliver splice-modulating oligonucleotides (phosphorodiamidate morpholino oligomers [PMOs]) to different target cell types.

MATERIALS & METHODS

EEV Library Screening and Optimization

- EEVs with robust target cell uptake and efficacy were screened in vitro, and functional validation was assessed. Well-tolerated EEVs with desired tissue functional delivery were then assessed in wild-type and disease animal models. Finally, an EEV candidate with desired therapeutic effect was selected.

DMD Studies

- DMD is caused by mutations in the *DMD* gene, resulting in the production of nonfunctional dystrophin protein (**Figure 2**).[3,4] Currently approved unconjugated PMO therapies were designed to restore the reading frame and produce dystrophin by exon skipping but have shown modest improvements.[5,6]
- EEV-PMO-23, a DMD exon 23 skipping PMO conjugated to the EEV platform, was administered intravenously to assess exon skipping and dystrophin production in D2-*mdx*[7] mice (**Figure 3**). These mice contain a nonsense mutation in exon 23.
- ENTR-601-44, a DMD exon 44 skipping PMO conjugated to the EEV platform, was administered to non-human primates (NHP) to assess exon skipping in cardiac and skeletal muscles (**Figure 4**).
- Exon skipping efficiency was analyzed by RT-PCR and LabChip. Dystrophin protein restoration in cells was evaluated by Simple Western Jess and immunofluorescence.

IRF5 Studies

- Interferon regulatory factor 5 (IRF5) is a transcription factor that promotes production of several proinflammatory cytokines in macrophages, and overexpression of IRF5 has been implicated in several autoimmune and inflammatory diseases.[8-11]
- IRF5 experiments used several EEVs conjugated to an exon skipping PMO.[12] The PMO in this conjugate facilitates exon 4 skipping of IRF5 mRNA that produces a premature stop codon, thereby reducing translation of IRF5 mRNA (**Figure 5A**).[12]

OBJECTIVES

- To evaluate the ability of the EEV platform to deliver PMOs in preclinical models of Duchenne muscular dystrophy (DMD) and murine macrophages.
- To evaluate functional ability of EEV-PMO conjugates to upregulate or downregulate target genes.

Figure 1. EEV-PMO Structure and Mechanism of Action.

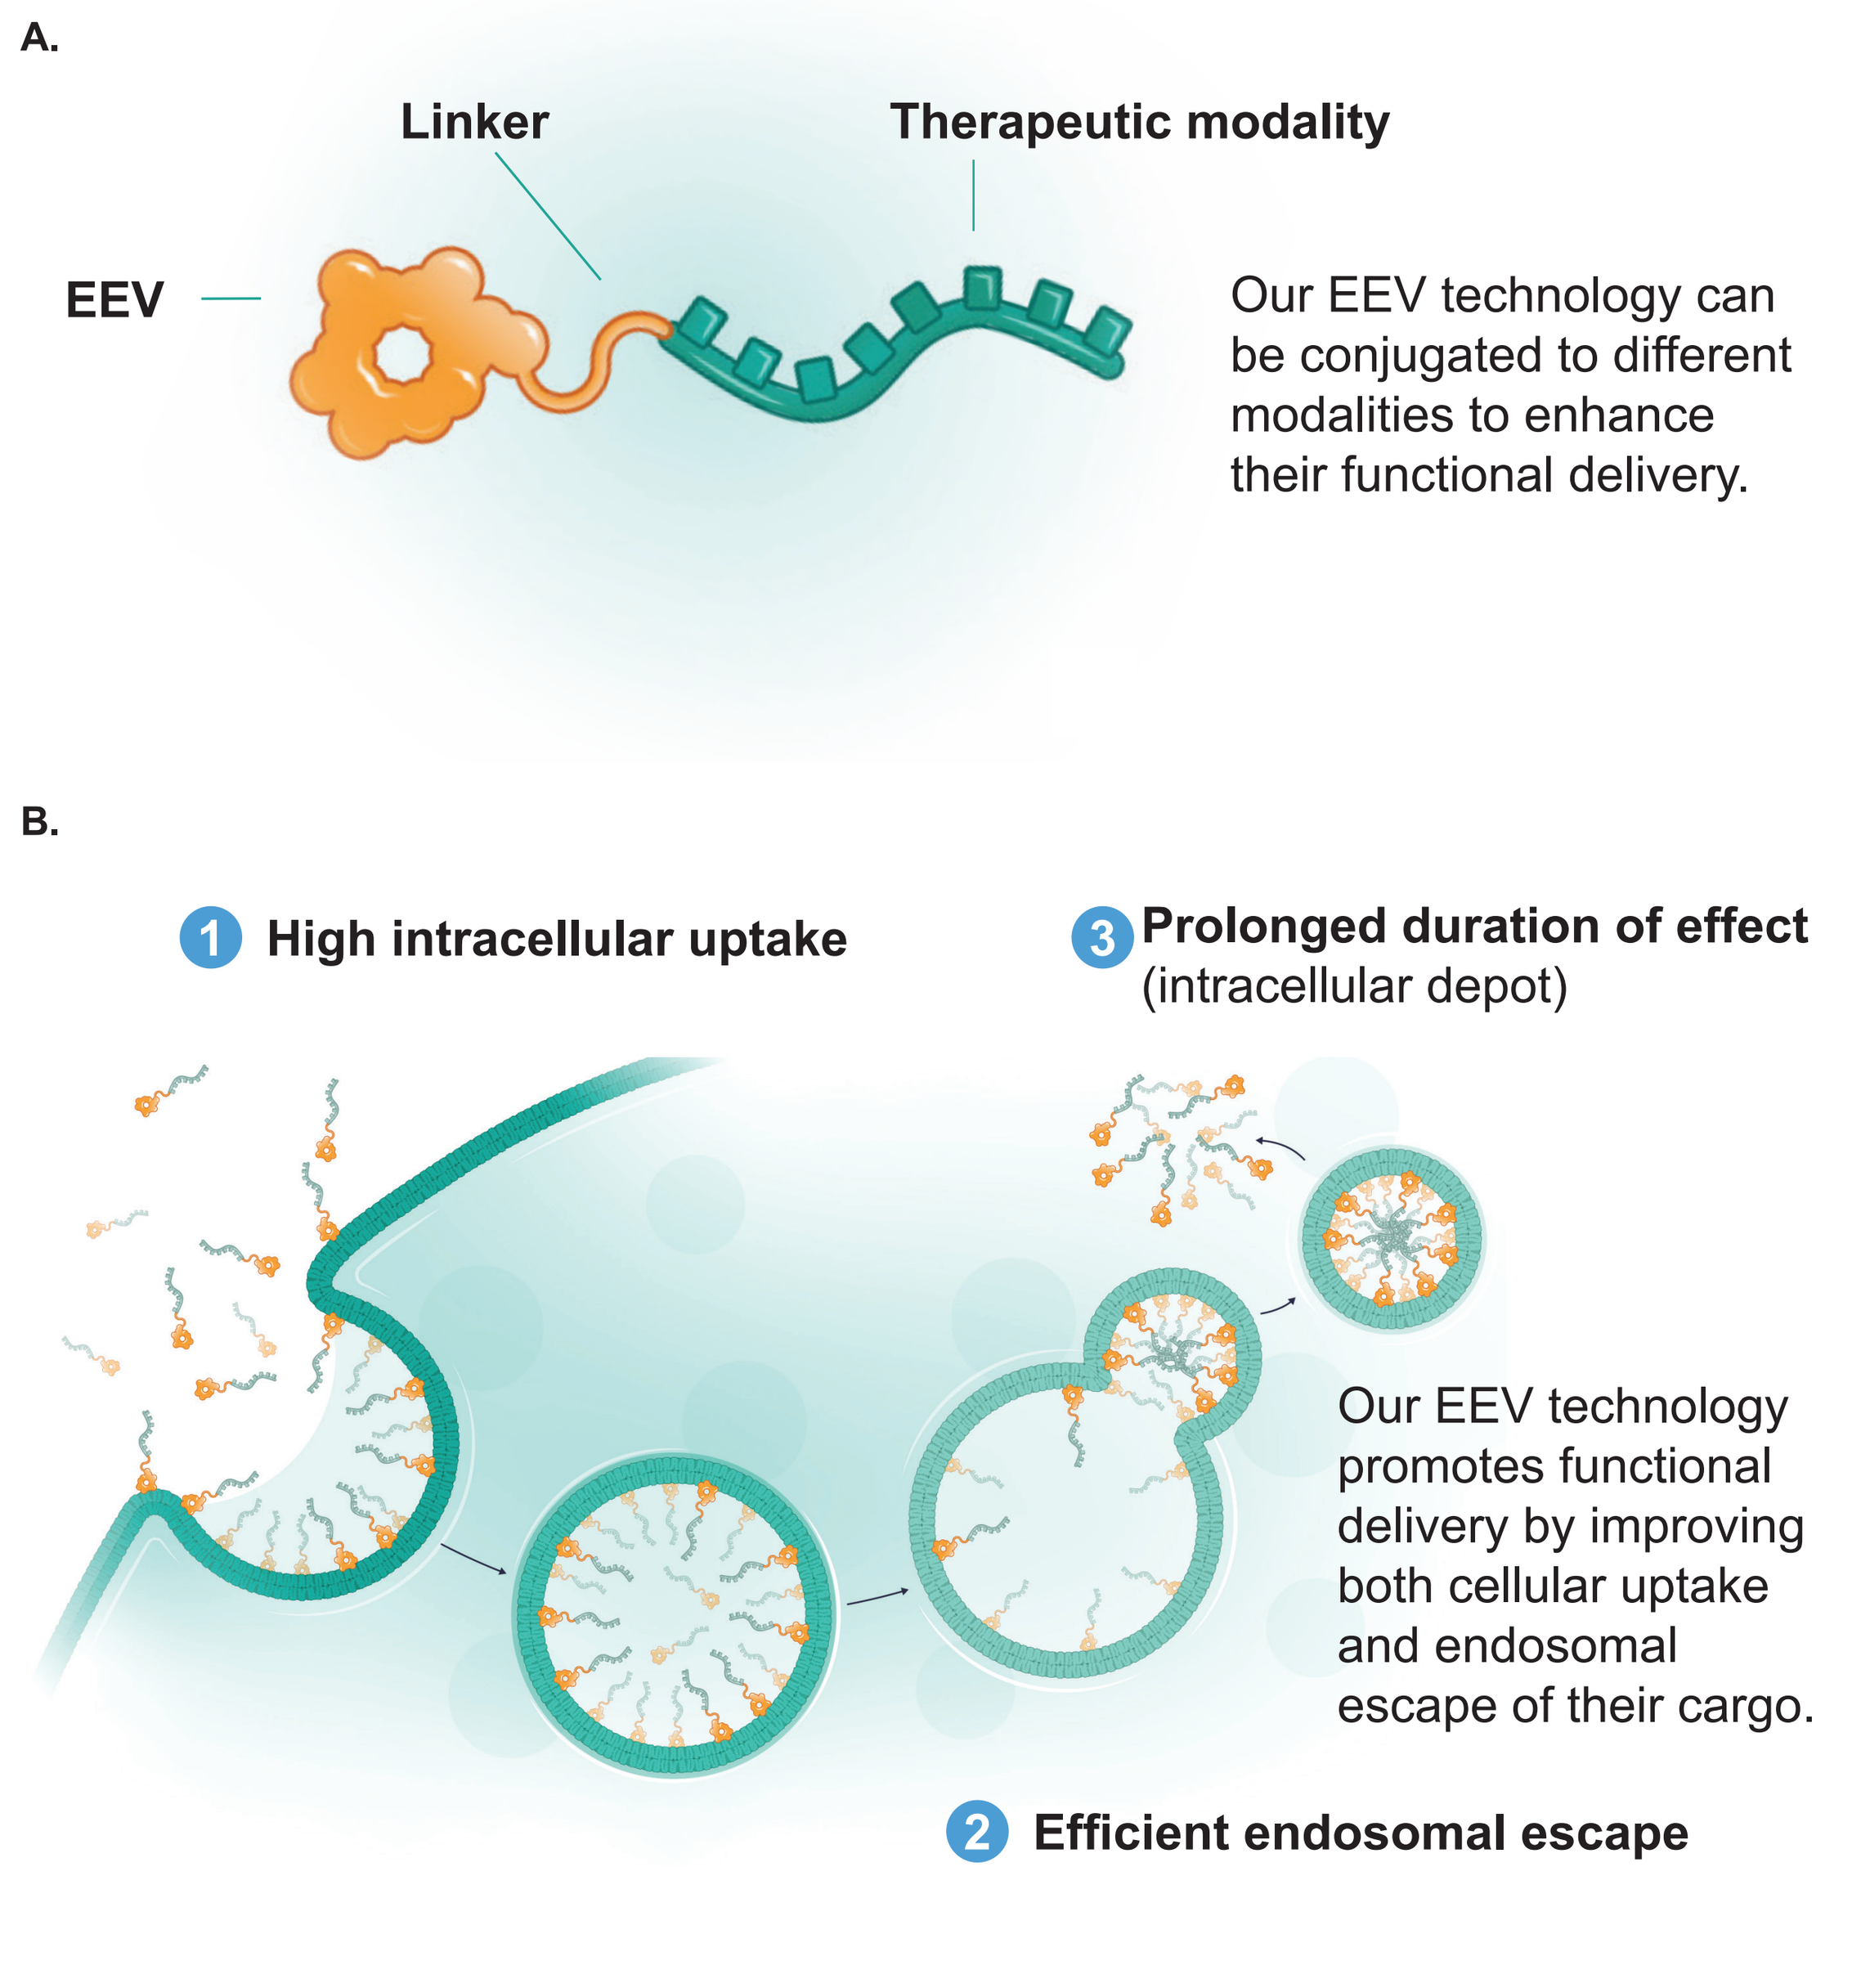
A.
Linker
Therapeutic modality
EEV
Our EEV technology can be conjugated to different modalities to enhance their functional delivery.
B.
1 High intracellular uptake
3 Prolonged duration of effect (intracellular depot)
Our EEV technology promotes functional delivery by improving both cellular uptake and endosomal escape of their cargo.
2 Efficient endosomal escape

(A) Structure of EEV-conjugated therapeutic consisting of cCPP-based EEV, linker, and therapeutic modality. **(B)** Mechanism of EEV-mediated delivery of therapeutics. EEV, endosomal escape vehicle; cCPP, cyclic cell-penetrating peptide.

RESULTS

Upregulation of Target Genes

Figure 2. Mechanism of EEV-PMO Conjugates in DMD Models.

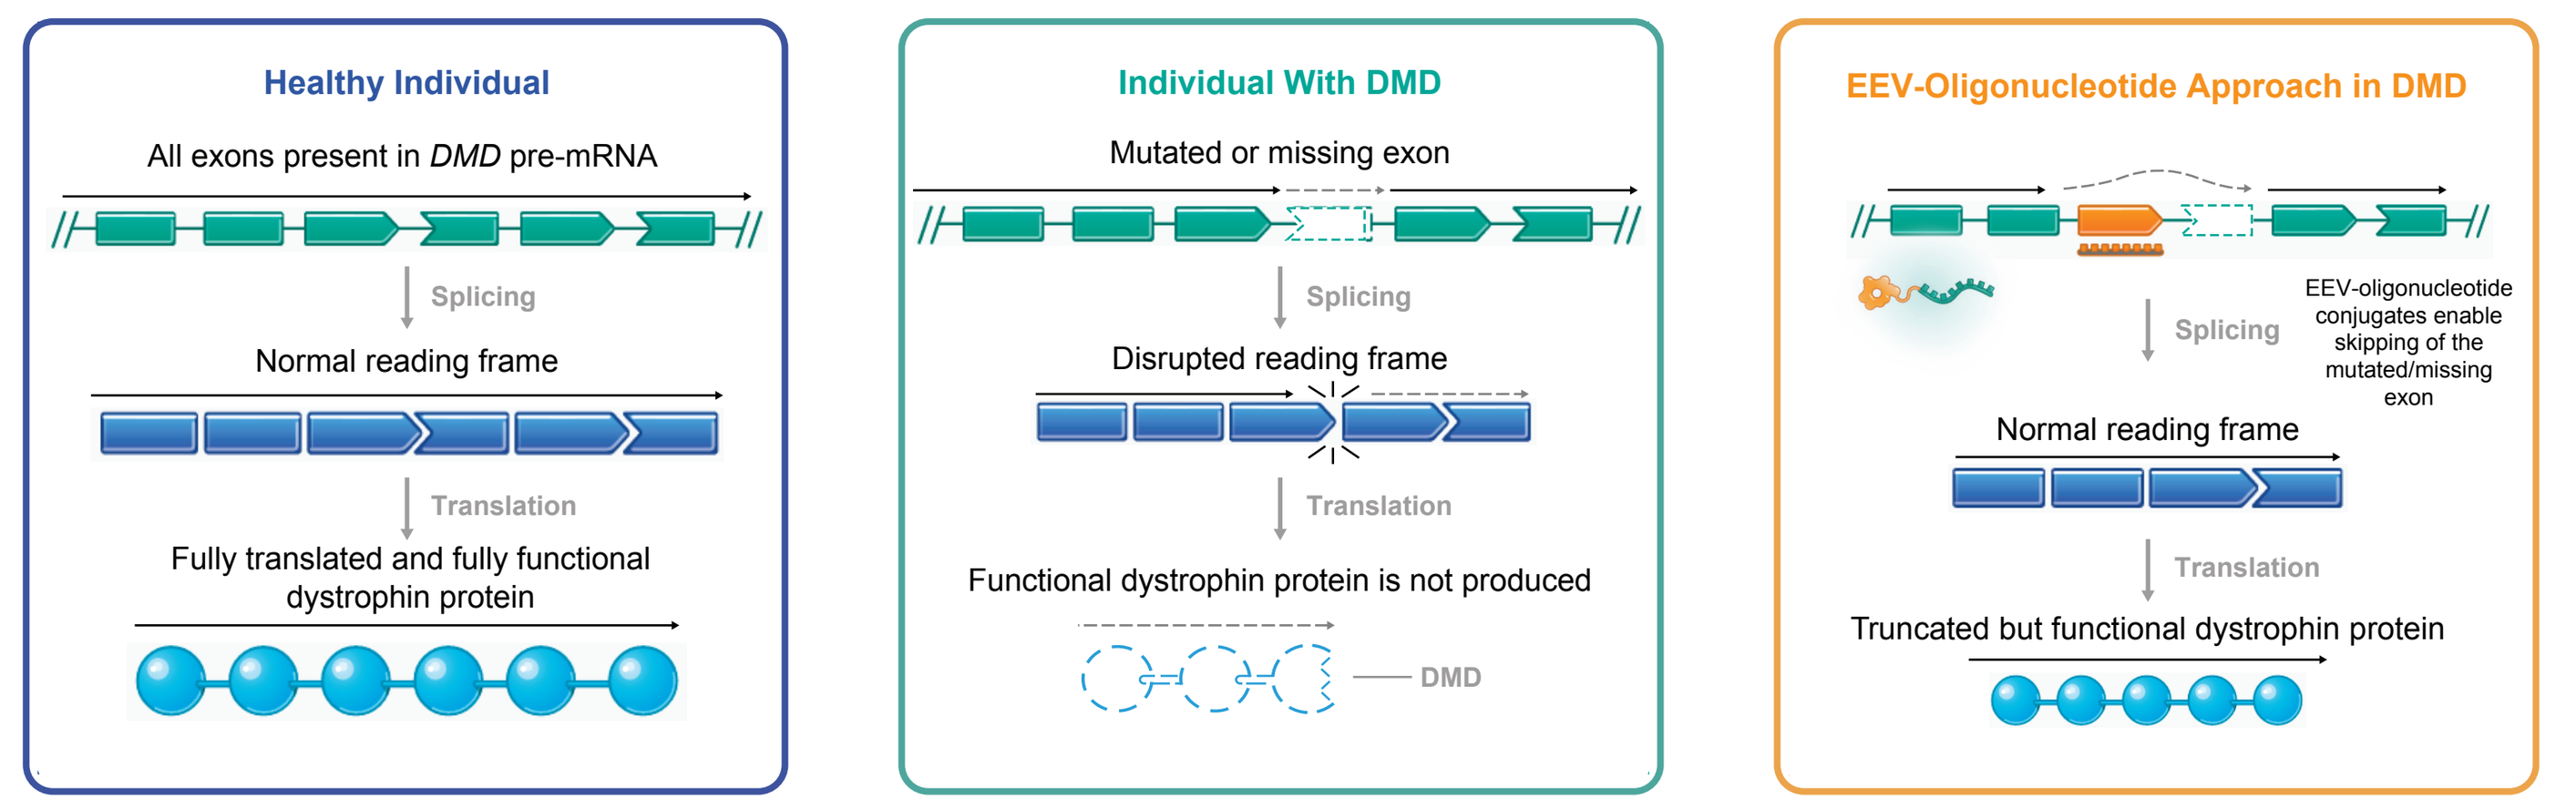
Healthy Individual
All exons present in DMD pre-mRNA
Splicing
Normal reading frame
Translation
Fully translated and fully functional dystrophin protein
Individual With DMD
Mutated or missing exon
Splicing
Disrupted reading frame
Translation
Functional dystrophin protein is not produced
DMD
EEV-Oligonucleotide Approach in DMD
Splicing
EEV-oligonucleotide conjugates enable skipping of the mutated/missing exon
Normal reading frame
Translation
Truncated but functional dystrophin protein

DMD, Duchenne muscular dystrophy; EEV, endosomal escape vehicle.

Exon Skipping and Dystrophin Restoration in D2-*mdx* Model of DMD

Broad dystrophin expression and restoration of skeletal and cardiac muscle integrity were observed with monthly EEV-PMO-23 administration compared with PMO-23 alone (**Figure 3A**). Bi-weekly treatment with EEV-PMO-23 improved skeletal muscle contractile force in D2-*mdx* mice which was not significantly different than wild-type mice at Week 12 (**Figure 3B**).

Figure 3. Repeat EEV-PMO Treatment and Improved Muscle Contractility in D2-*mdx* Mice.

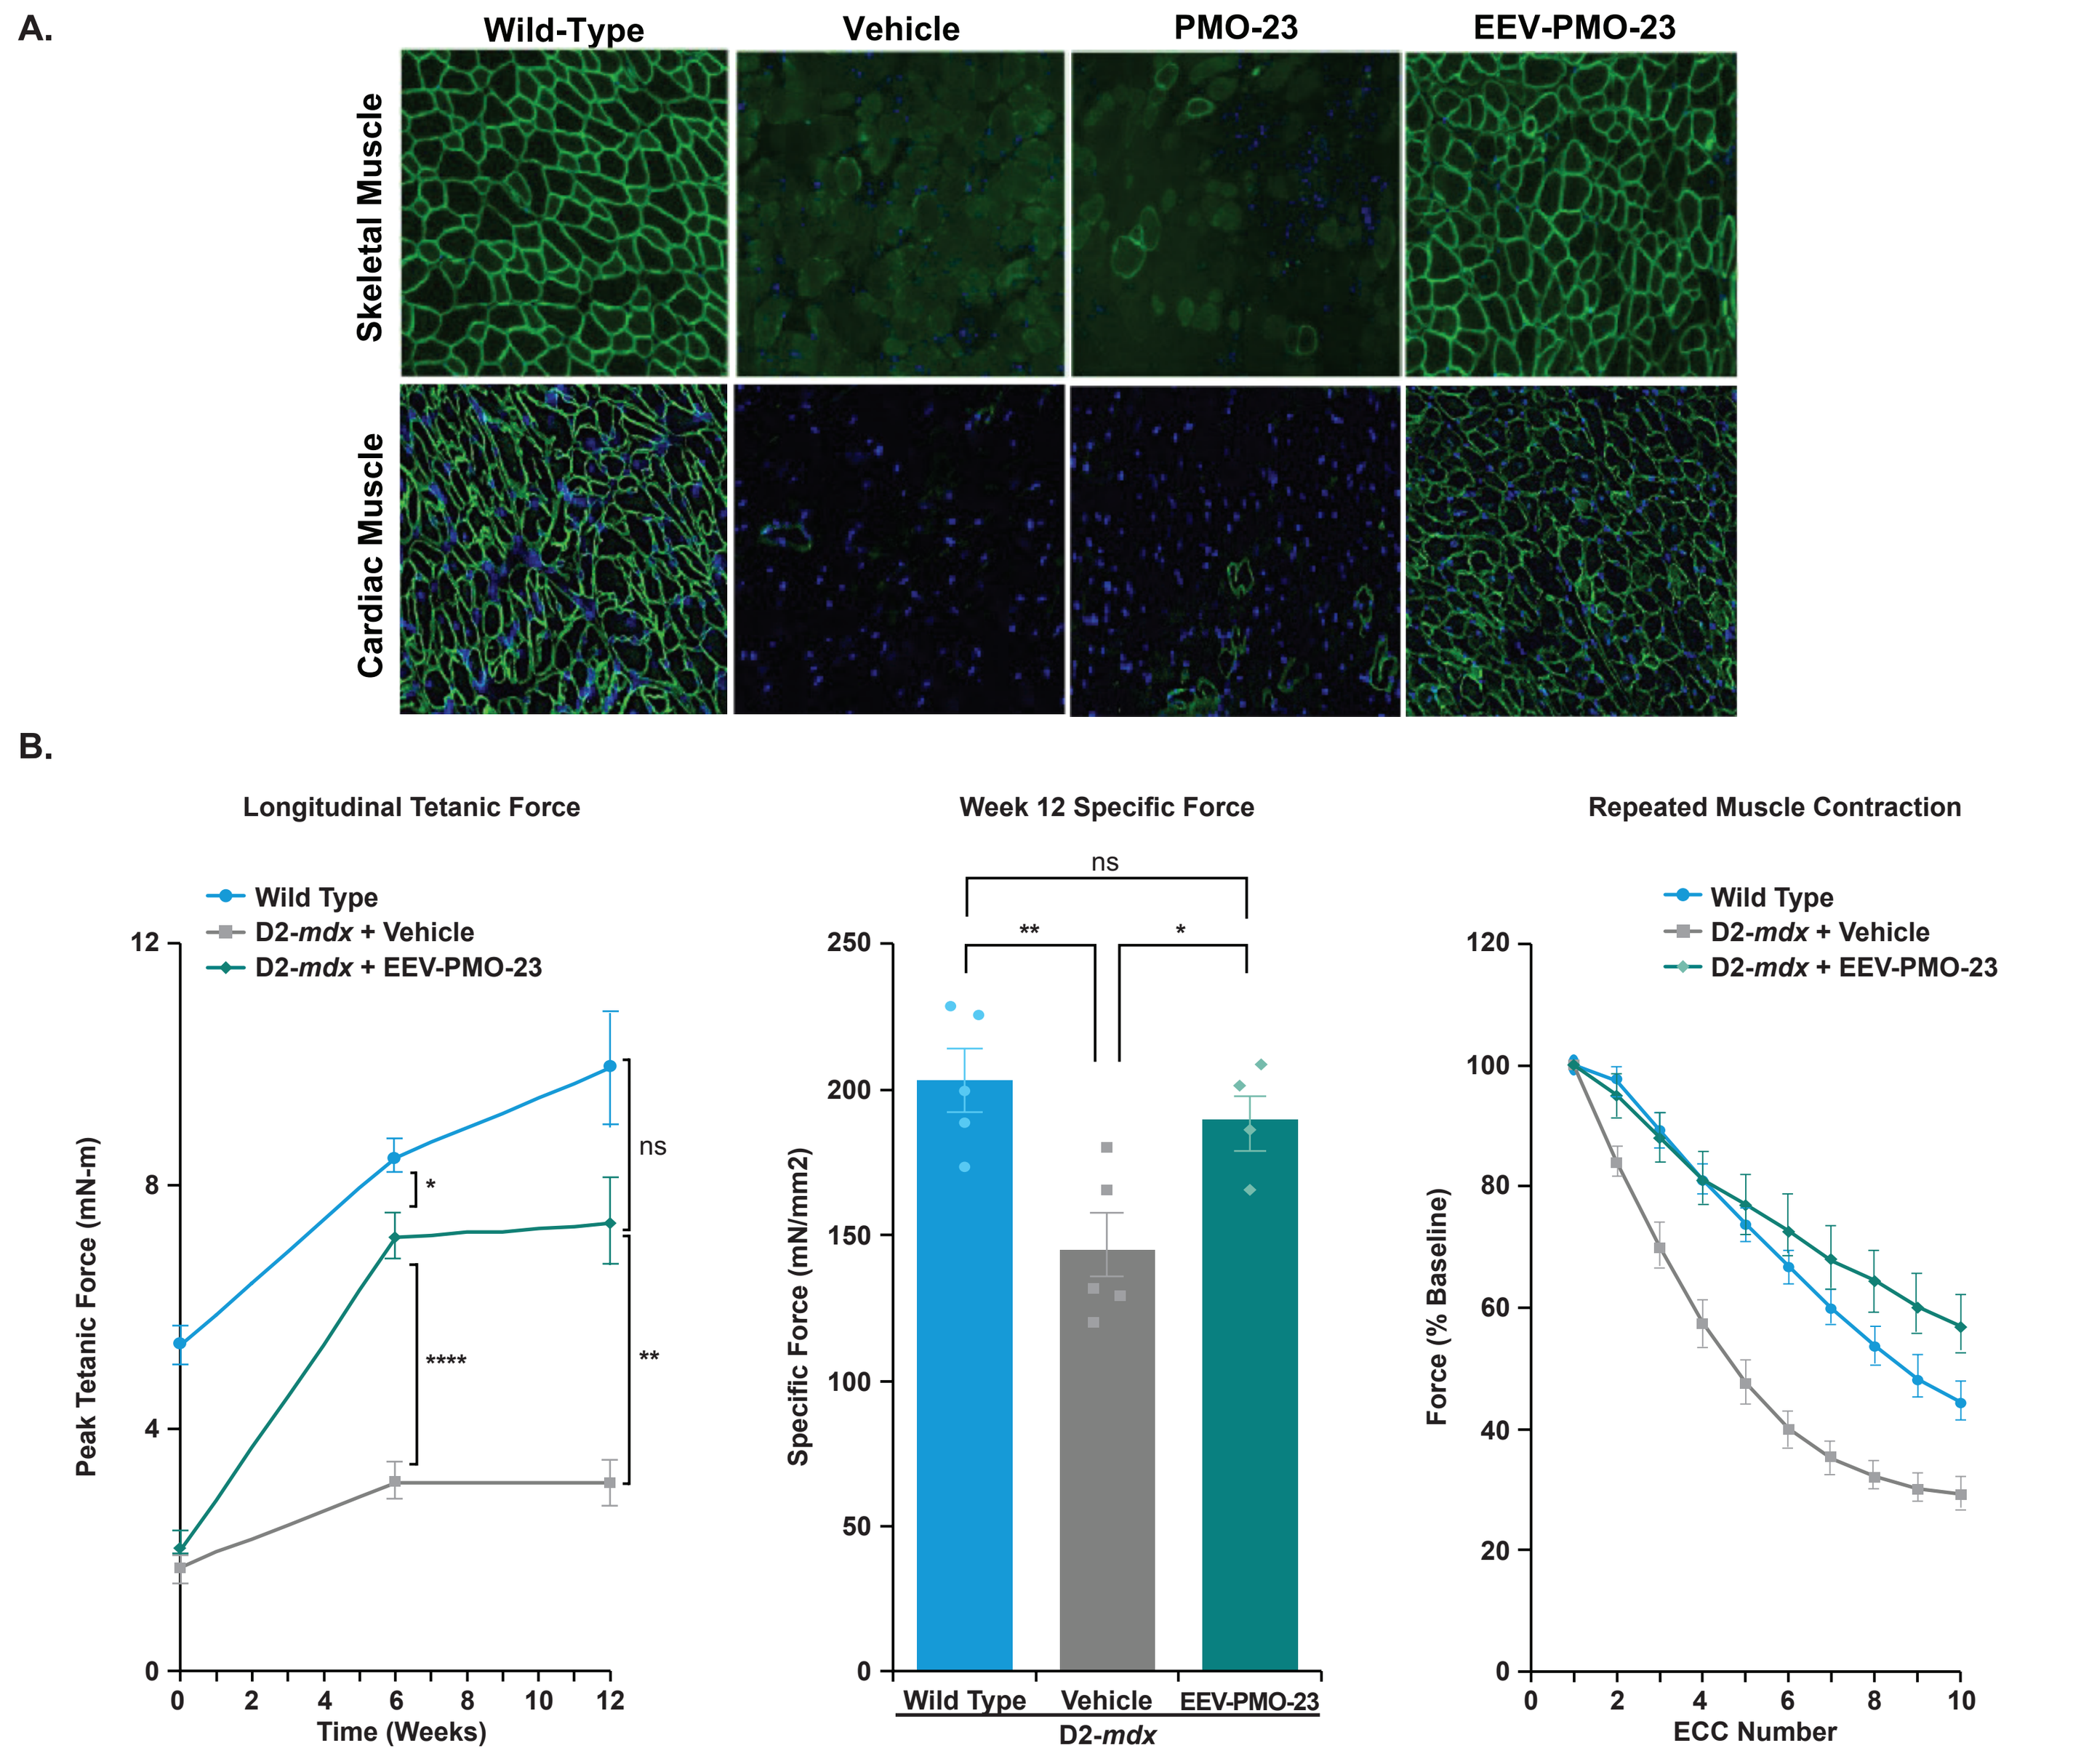
A.
Wild-Type
Vehicle
PMO-23
EEV-PMO-23
Skeletal Muscle
Cardiac Muscle
B.
Longitudinal Tetanic Force
Wild Type
D2-mdx + Vehicle
D2-mdx + EEV-PMO-23
Peak Tetanic Force (mN-m)
Time (Weeks)
ns
*

**
Week 12 Specific Force
Specific Force (mN/mm2)
Wild Type
Vehicle
EEV-PMO-23
D2-mdx
Repeated Muscle Contraction
Force (% Baseline)
ECC Number

(A) D2-*mdx* mice (male, n=6-7) were treated with 4 monthly doses of either vehicle, 20 mg/kg PMO or 20 mg/kg PMO equivalent of EEV-PMO, and the data were collected ~4 weeks after the last dose. Dystrophin in green and DAPI in blue. **(B)** D2-*mdx* mice were treated with vehicle or 80 mg/kg (Q2W) EEV-PMO-23. Muscle contractility was assessed at Weeks 6 and 12 via isometric force generated by tetanic contraction of plantar flexor muscle group and ECC generated by repeated tetanic contraction of the tibialis anterior muscle. *p<0.05, **p<0.01, ****p<0.0001; ns, not significant; shown as mean ± SEM. DAPI, 4′,6-diamidino-2-phenylindole; EEC, eccentric force; EEV, endosomal escape vehicle; PMO, phosphorodiamidate morpholino oligomer; Q2W, once every 2 weeks.

Durable Efficacy of ENTR-601-44 in NHP Models of DMD

A single dose of ENTR-601-44 resulted in robust exon skipping in both skeletal and cardiac muscles in NHPs, as well as prolonged duration of effect for at least 12 weeks (**Figure 4**).

Figure 4. ENTR-601-44 in NHPs.

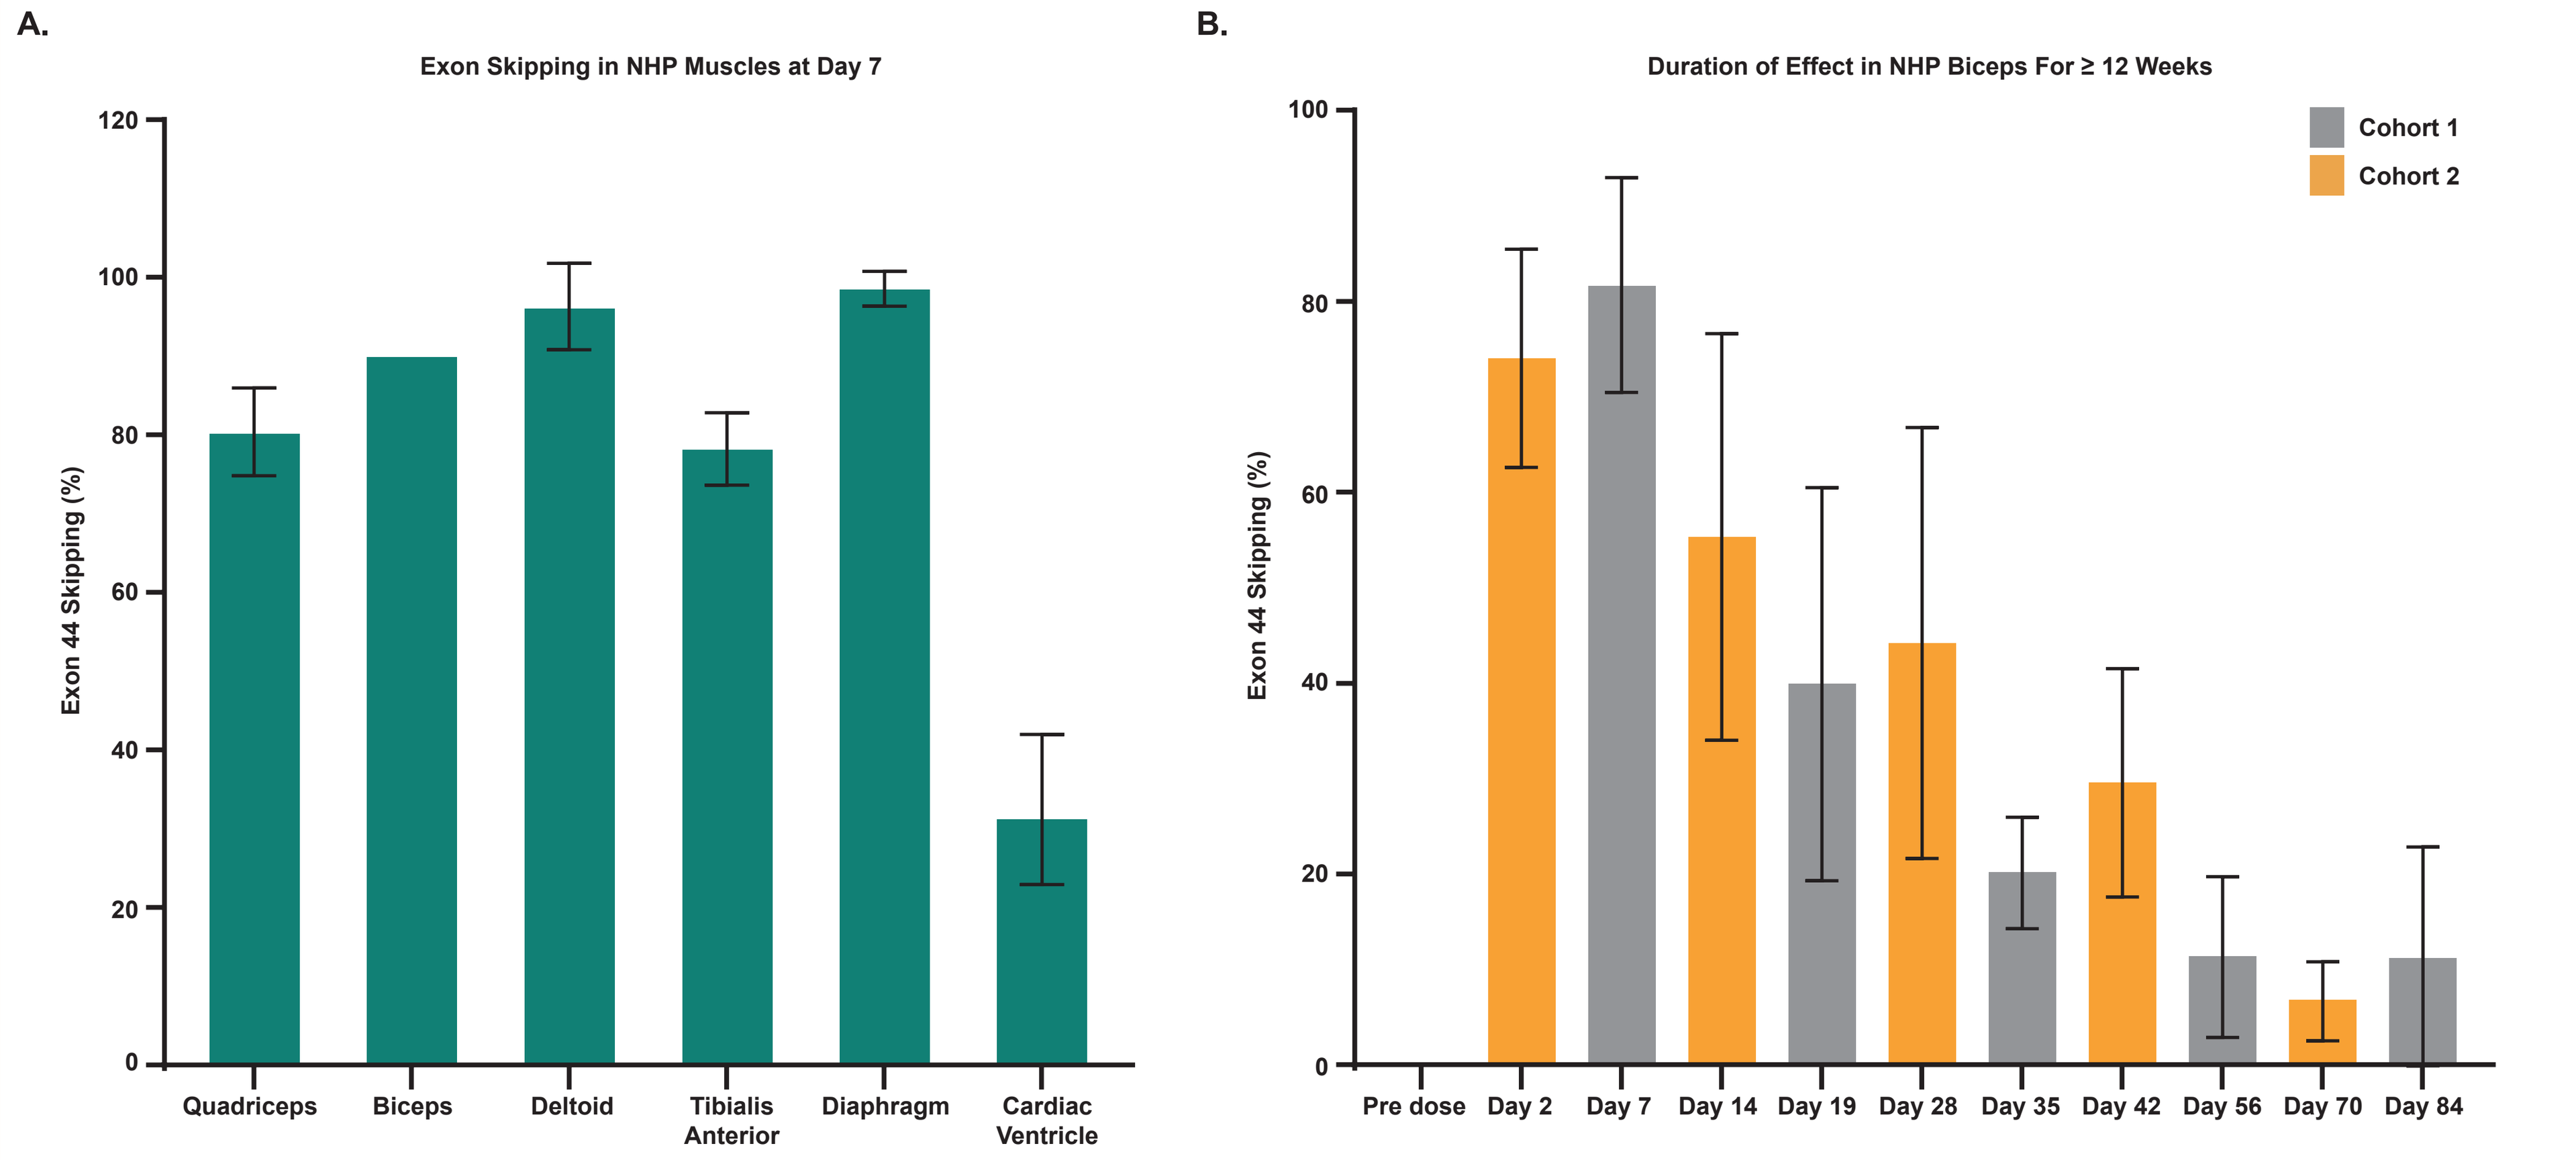
A.
Exon Skipping in NHP Muscles at Day 7
Exon 44 Skipping (%)
Quadriceps
Biceps
Deltoid
Tibialis Anterior
Diaphragm
Cardiac Ventricle
B.
Duration of Effect in NHP Biceps For ≥ 12 Weeks
Cohort 1
Cohort 2
Exon 44 Skipping (%)
Pre dose
Day 2
Day 7
Day 14
Day 19
Day 28
Day 35
Day 42
Day 56
Day 70
Day 84

(A) At 7 days post-IV infusion of 30 mg/kg (PMO equivalent), robust exon 44 skipping across different muscle groups isolated from the ENTR-601-44–treated NHP. **(B)** Post-intravenous infusion of 35 mg/kg (PMO equivalent), robust exon 44 skipping observed in biceps in the ENTR-601-44–treated NHP (n=3 per cohort) for ≥12 weeks. NHP, non-human primate; PMO, phosphorodiamidate morpholino oligomer.

Downregulation of Target Genes

EEV-PMO Treatment Reduced IRF5 Expression In Vitro and In Vivo

Four different EEV-PMO conjugates reduced IRF5 expression in mouse macrophages stimulated to produce IRF5 (**Figure 5B**). EEV#1-PMO also induced downregulation of IRF5 in liver, small intestine, and skeletal muscle of wild-type mice.

Figure 5. EEV-PMO Knockdown of IRF5 Expression.

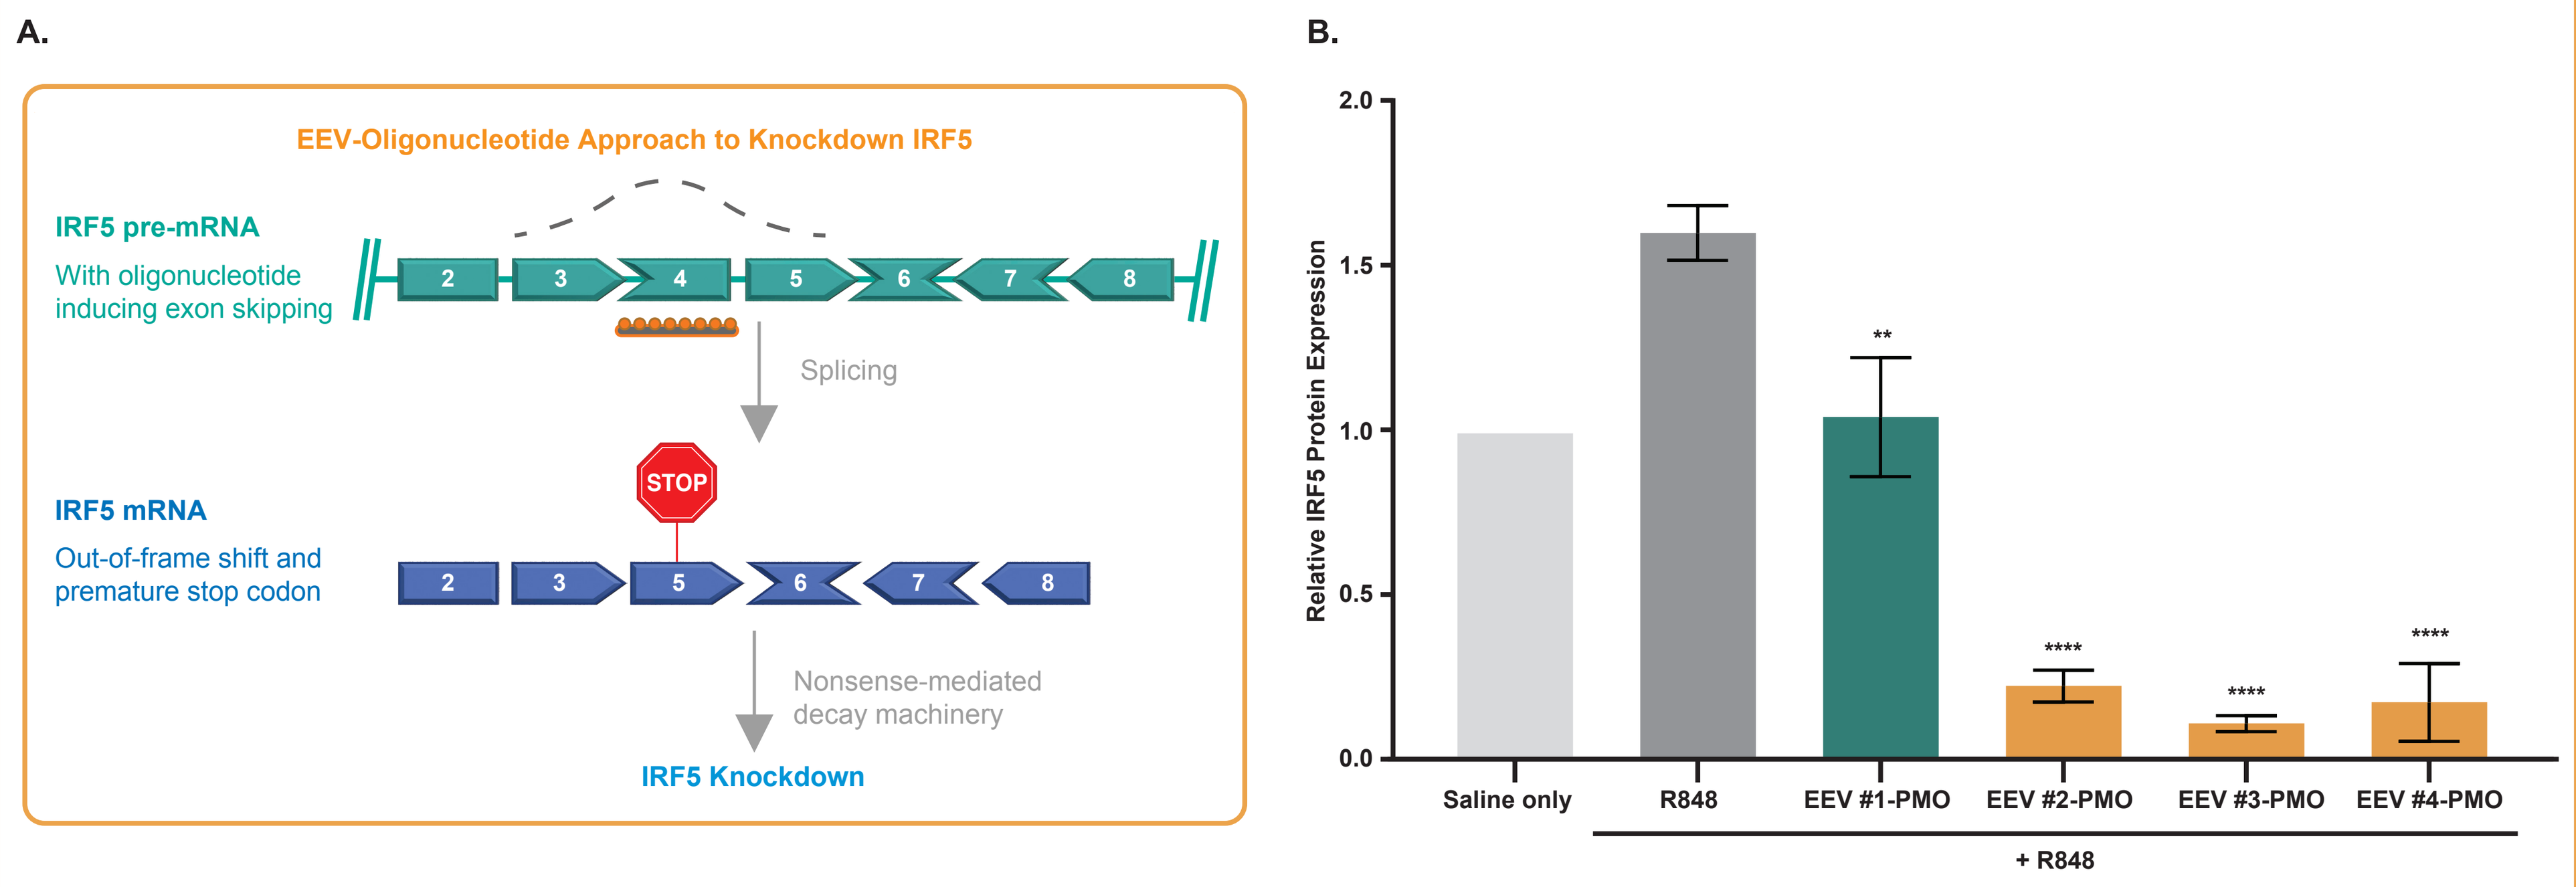
A.
EEV-Oligonucleotide Approach to Knockdown IRF5
IRF5 pre-mRNA
With oligonucleotide inducing exon skipping
Splicing
STOP
IRF5 mRNA
Out-of-frame shift and premature stop codon
Nonsense-mediated decay machinery
IRF5 Knockdown
B.
Relative IRF5 Protein Expression
Saline only
R848
EEV #1-PMO
EEV #2-PMO
EEV #3-PMO
EEV #4-PMO
+ R848
**

(A) Mechanism of EEV#1-PMO for IRF5 knockdown. **(B)** Mouse macrophage cells were pre-treated with different EEV (#1-4) conjugated to PMO at 2 micromolar for 4 hours, followed by stimulation with R848 to stimulate IRF5 production overnight. At 24 hours post treatment, cells were harvested and evaluated by Western Blot. R848 is a TLR7/8 agonist that upregulates IRF5. **p<0.01, ****p<0.0001 versus R848-treated cells. EEV, endosomal escape vehicle; IRF5, interferon regulatory factor 5; PMO, phosphorodiamidate morpholino oligomer; TLR, toll-like receptor.

CONCLUSIONS

- The EEV™ platform consists of a library of proprietary cyclic peptides with unique chemistry that enable improved cellular uptake, endosomal escape, and consistent translation across species.
- The results presented here demonstrate that the EEV platform successfully and efficiently delivers oligonucleotides to specific cell and tissue types.
- The EEV-PMO approach has broad applicability to upregulate or downregulate target genes through distinct mechanisms of action.

ACKNOWLEDGEMENTS

This research was funded by Entrada Therapeutics, Inc. (Boston, MA). Vlad Batagui and Suresh Peddigari (formerly at Entrada) contributed to this research. Editorial and graphics support for the development of this poster was provided by Ashfield MedComms, an Inizio Company, and funded by Entrada Therapeutics, Inc. References: 1. Qian Z. *Biochemistry*. 2016. 2. Sahni A. *ACS Chem Biol*. 2020. 3. Hoffman EP. *Cell*. 1987. 4. Birnkrant DJ. *Lancet Neurol*. 2018. 5. EXONDYS 51® Prescribing Information. 6. VILTEPSO® Prescribing Information. 8. Takaoka A. *Nature*. 2005. 9. Graham RR. *Nat Genet*. 2006. 10. Krausruber T. *Nat Immunol*. 2011. 11. Yang Y. *Inflamm Bowel Dis*. 2021. 12. Data on File, Entrada Therapeutics, Inc.

26th Annual Meeting of the American Society of Gene & Cell Therapy (ASGCT)

Entrada Therapeutics, Inc., Boston, MA.