Exon Skipping and Dystrophin Production With ENTR-601-45, a Novel EEV-Conjugated, Exon 45 Skip-Amenable PMO in Preclinical Models of DMD

Xiang Li, Nelsa L. Estrella, Mark Wysk, Mahboubeh Kheirabadi, Patrick G. Dougherty, Ajay Kumar, Amy N. Hicks, Anushree Pathak, Avery Guan, Sara L. Blake, Se Yun Cheon, Christopher M. Brennan, Wenlong Lian, Nanjun Liu, Matthew Streeter, Andy Stadheim, Mohanraj Dhanabal, Mahasweta Girgenrath, Ziqing Leo Qian

Entrada Therapeutics, Boston, MA

Poster #1348

INTRODUCTION

- Duchenne muscular dystrophy (DMD) is an X-linked, progressive disease caused by mutations in the *DMD* gene, resulting in the production of nonfunctional dystrophin protein.[1-2]
- Currently approved phosphorodiamidate morpholino oligomer (PMO) therapies were designed to restore the reading frame and produce dystrophin by exon skipping **(Figure 1)**, but have shown modest improvements.[3-4]
 - Limited exposure and poor endosomal escape in target tissue after systemic administration of antisense oligonucleotides remain significant hurdles for the treatment of patients with DMD and result in insufficient dystrophin protein restoration, especially in cardiac muscle.
- To improve cellular uptake and enhance endosomal escape of oligonucleotides and other biologics, we developed an Endosomal Escape Vehicle (EEV) delivery platform based on cyclic cell-penetrating peptides (cCPPs).[5-6]
- We previously demonstrated that D2-*mdx* mice treated with EEV-PMO-23 produced durable and robust exon 23 skipping and dystrophin production in both skeletal and cardiac muscle. Additionally, ENTR-601-44, a clinical candidate for exon 44 skip-amenable patients, produced robust exon 44 skipping in hDMD mice, and showed both robust exon skipping and dystrophin protein restoration in patient-derived skeletal muscle cells.[7]
- We developed ENTR-601-45 for the treatment of exon 45 skip-amenable mutations in DMD patients to further address unmet medical needs.

MATERIALS & METHODS

- ENTR-601-45 is a *DMD* exon 45 skip-amenable PMO conjugated to the EEV platform.
- ENTR-601-45 efficacy was assessed in iPSC-derived skeletal and cardiac muscle cells from an exon 45 skip-amenable DMD patient harboring an exon 46-48 deletion mutation. Myotubes were treated for 24 hours and analyzed after 5 days of differentiation. Cardiomyocytes were treated for 24 hours and analyzed 48 hours later.
- Exon skipping efficiency was analyzed by RT-PCR and LabChip. Dystrophin protein restoration in cells was evaluated by Simple Western Jess and immunofluorescence.
- Humanized DMD (hDMD)[8] mice expressing the full-length human *DMD* gene were treated intravenously with ENTR-601-45 to evaluate exon 45 skipping efficacy in vivo.

OBJECTIVES

- Assess the therapeutic potential of ENTR-601-45 for patients with exon 45 skip-amenable DMD in cell and animal models.

Figure 1. Mechanism of Exon Skipping With EEV-PMO Conjugates.

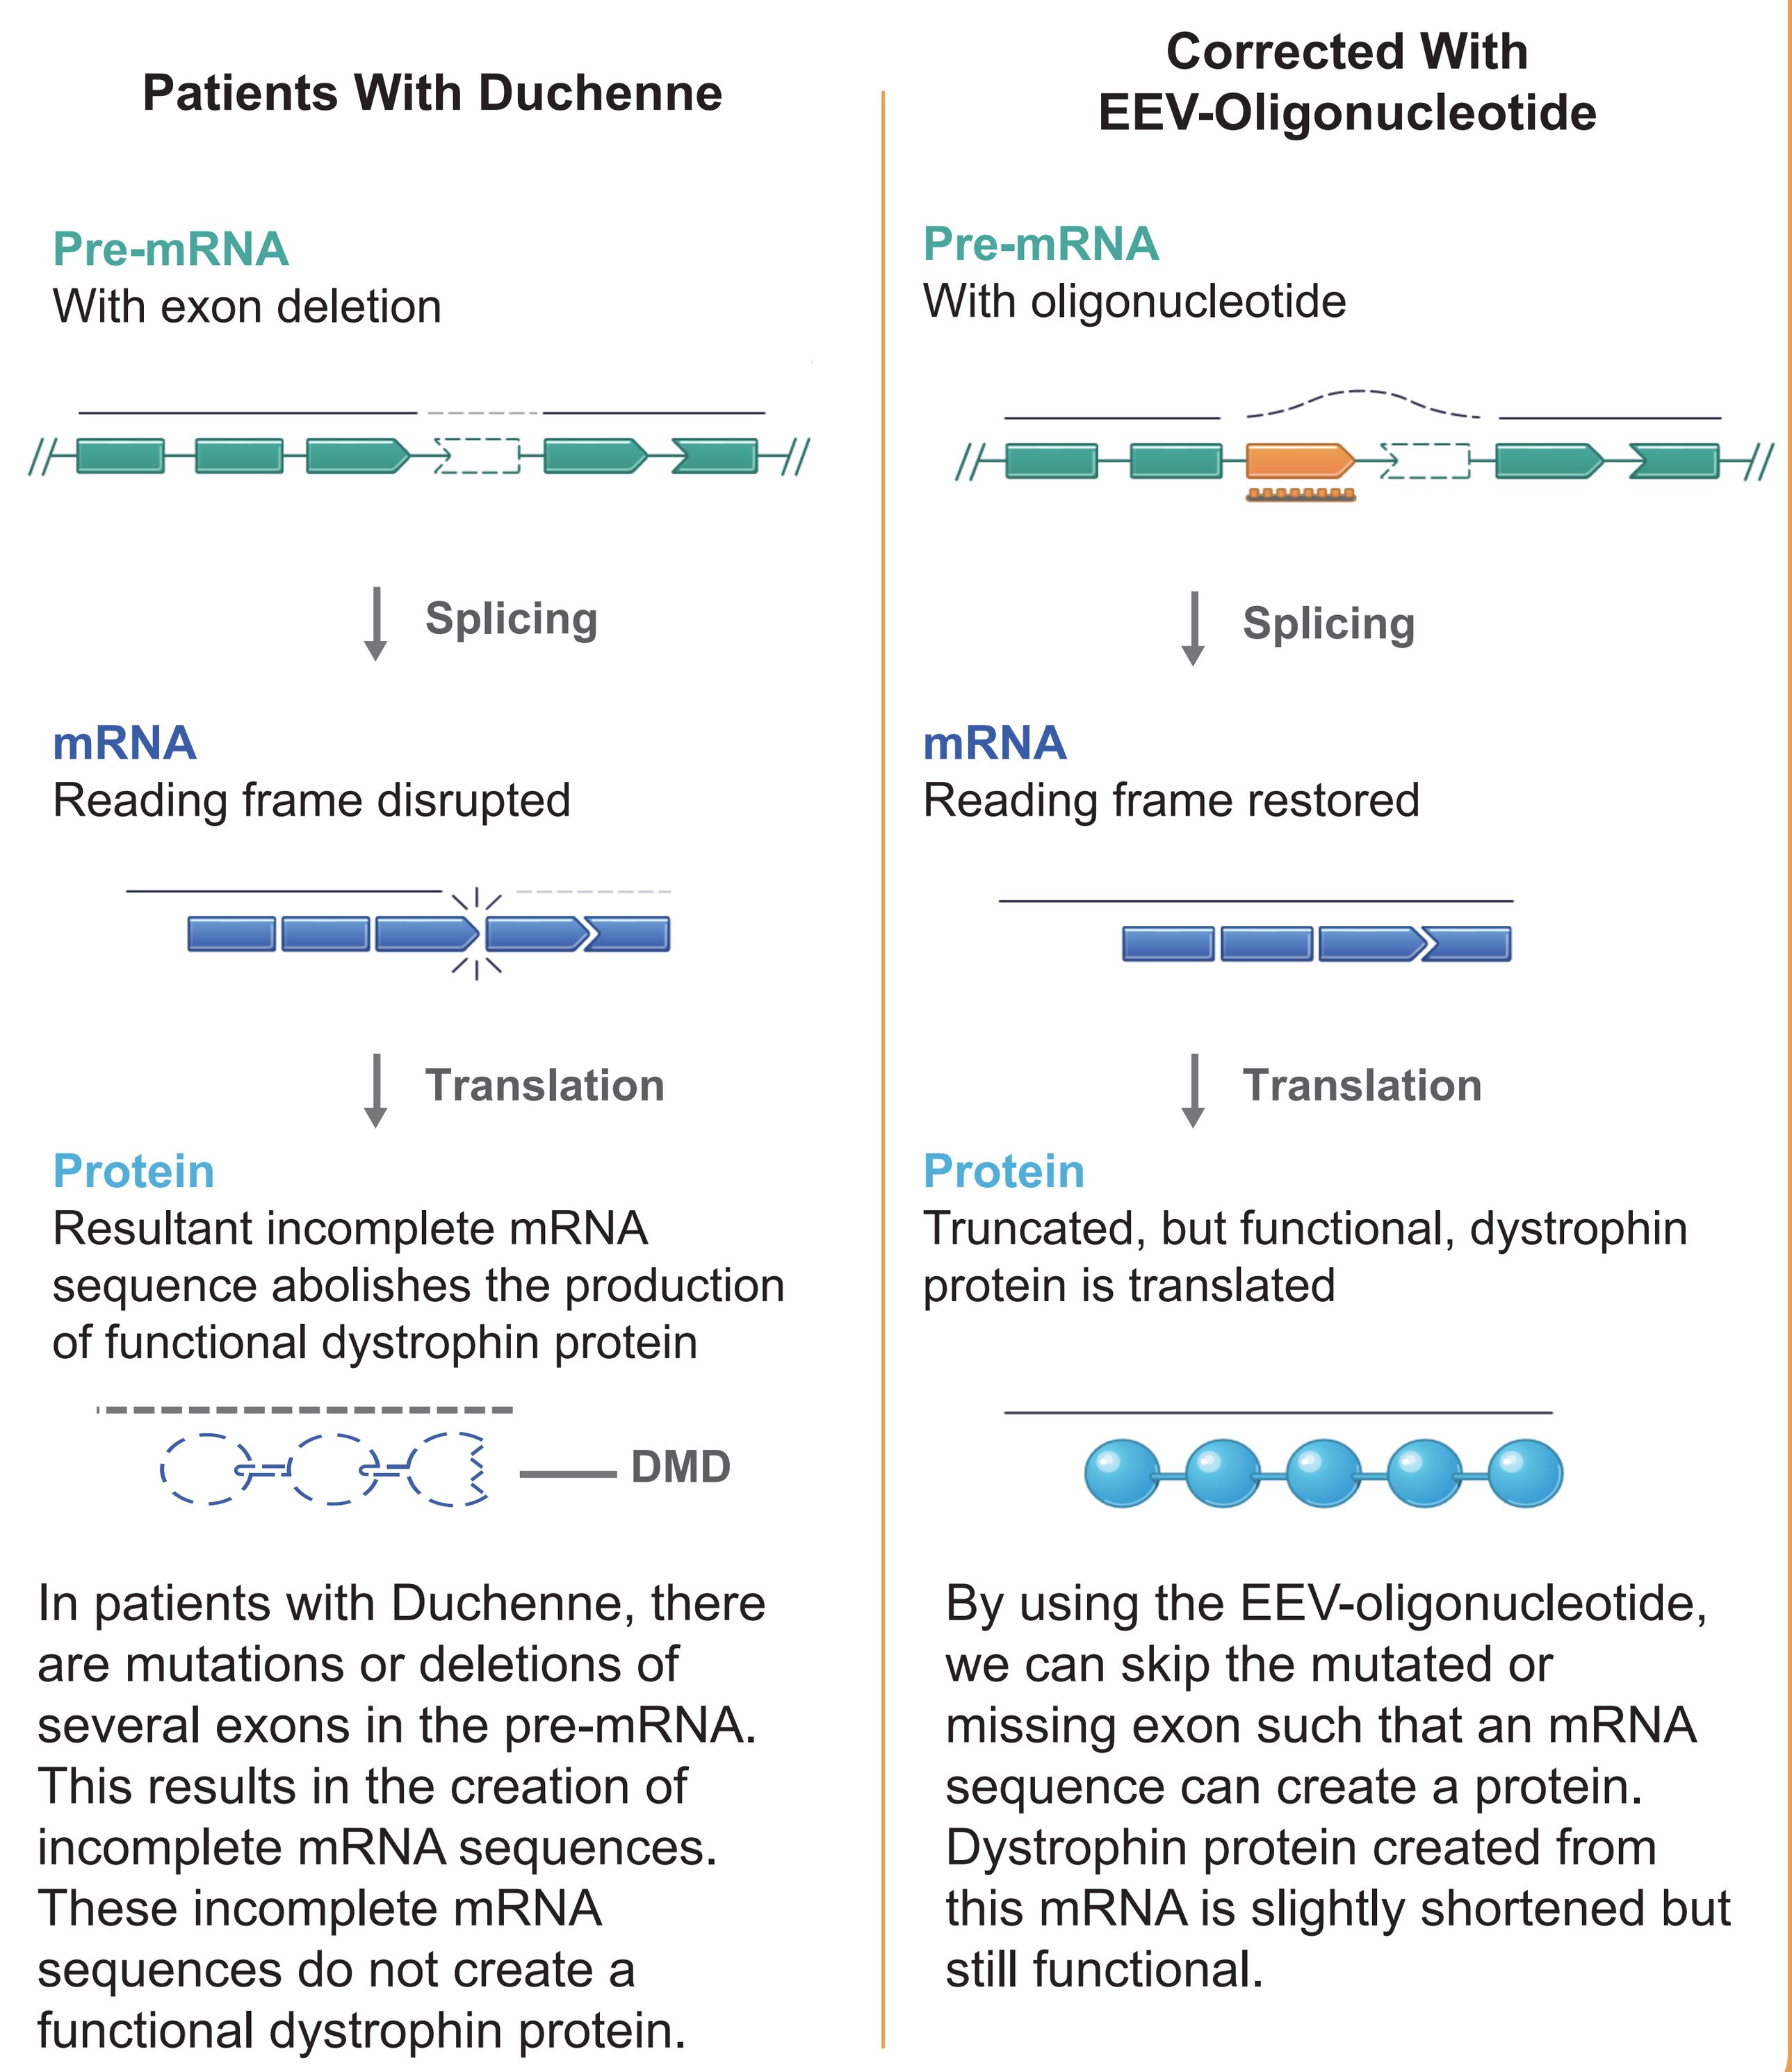
Patients With Duchenne
Pre-mRNA
With exon deletion
Splicing
mRNA
Reading frame disrupted
Translation
Protein
Resultant incomplete mRNA sequence abolishes the production of functional dystrophin protein
DMD
In patients with Duchenne, there are mutations or deletions of several exons in the pre-mRNA. This results in the creation of incomplete mRNA sequences. These incomplete mRNA sequences do not create a functional dystrophin protein.
Corrected With EEV-Oligonucleotide
Pre-mRNA
With oligonucleotide
Splicing
mRNA
Reading frame restored
Translation
Protein
Truncated, but functional, dystrophin protein is translated
By using the EEV-oligonucleotide, we can skip the mutated or missing exon such that an mRNA sequence can create a protein. Dystrophin protein created from this mRNA is slightly shortened but still functional.

RESULTS

In Vitro Exon Skipping and Dystrophin Restoration

ENTR-601-45 showed robust exon skipping and dystrophin production in patient-derived skeletal (**Figure 2**) and cardiac (**Figure 3**) muscle cells.

Figure 2. Efficacy of ENTR-601-45 in Skeletal Muscle Cells.

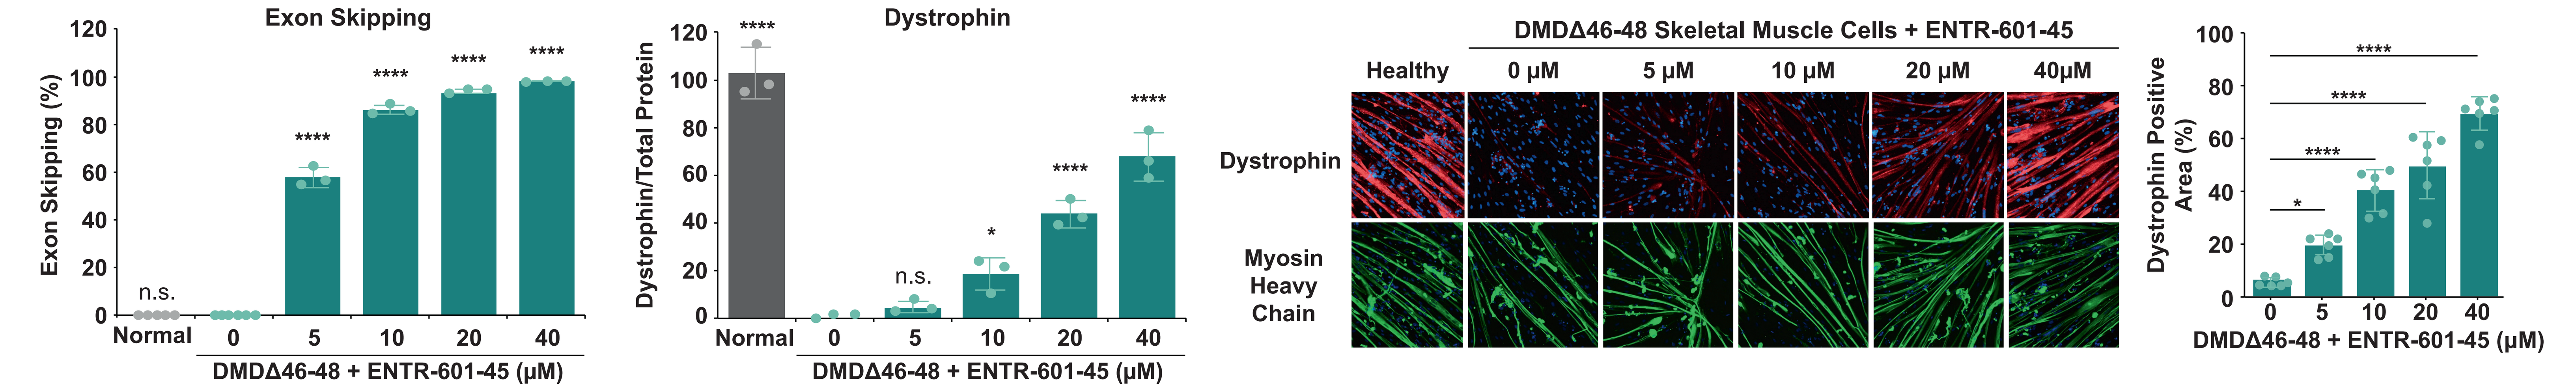
Exon Skipping
Exon Skipping (%)
Normal 0 5 10 20 40
DMDΔ46-48 + ENTR-601-45 (μM)
Dystrophin
Dystrophin/Total Protein
DMDΔ46-48 Skeletal Muscle Cells + ENTR-601-45
Healthy 0 μM 5 μM 10 μM 20 μM 40μM
Dystrophin
Myosin Heavy Chain
Dystrophin Positive Area (%)

DMDΔ46-48 patient-derived cells were incubated with ENTR-601-45. Data are shown as mean ± SD (n=3); one-way ANOVA; *p<0.05, ****p<0.0001; n.s., not significant; relative to untreated DMDΔ46-48 cells.

DMDΔ46-48 patient-derived skeletal muscle cells were incubated with ENTR-601-45. Data are shown as mean ± SD (n=6). Ordinary one-way ANOVA and Dunnett's multiple comparison test; *p<0.05****p<0.0001 relative to untreated DMDΔ46-48.

Figure 3. Efficacy of ENTR-601-45 in Cardiac Muscle Cells.

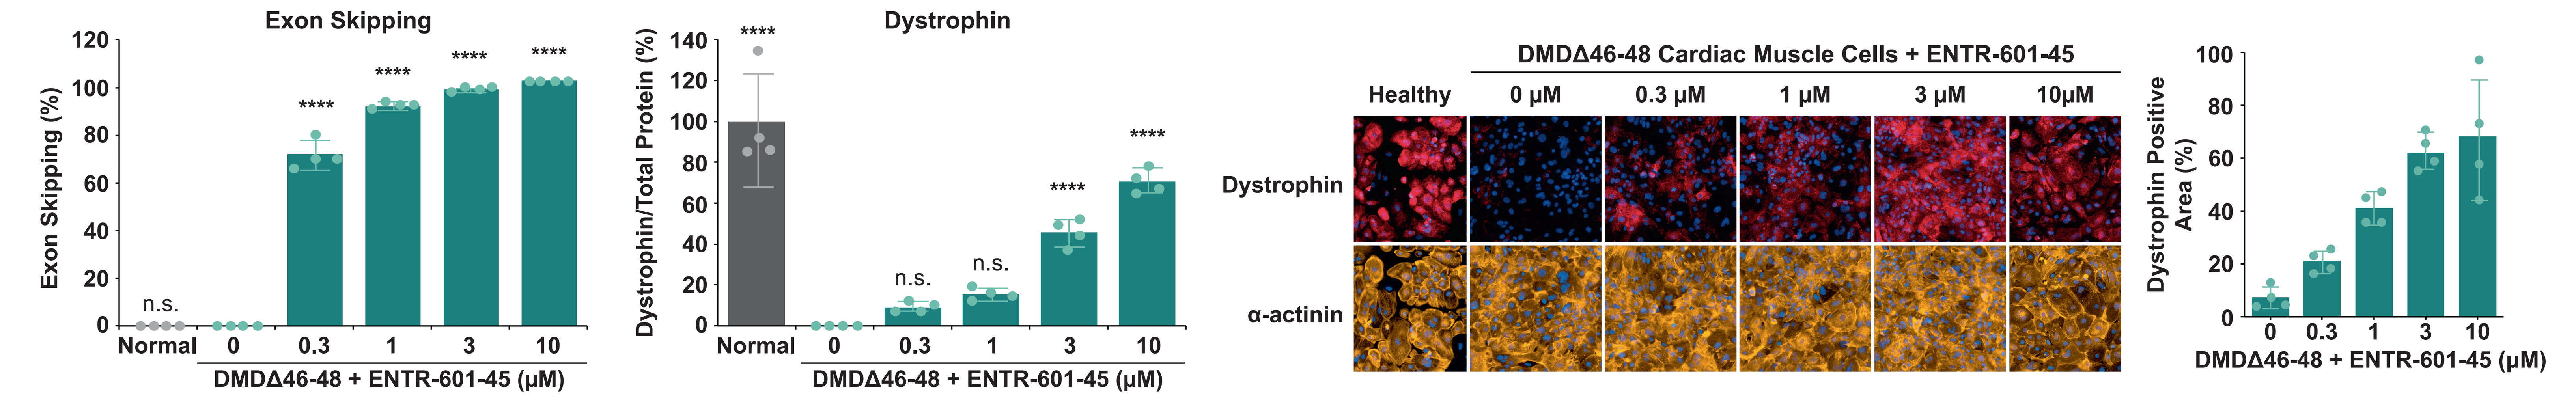
Exon Skipping
Exon Skipping (%)
Normal 0 0.3 1 3 10
DMDΔ46-48 + ENTR-601-45 (μM)
Dystrophin
Dystrophin/Total Protein (%)
DMDΔ46-48 Cardiac Muscle Cells + ENTR-601-45
Healthy 0 μM 0.3 μM 1 μM 3 μM 10μM
Dystrophin
α-actinin
Dystrophin Positive Area (%)

DMDΔ46-48 patient-derived cells were incubated with ENTR-601-45. Data are shown as mean ± SD (n=4); one-way ANOVA; ****p<0.0001; n.s., not significant; relative to untreated DMDΔ46-48 cells.

DMDΔ46-48 patient-derived cardiac muscle cells were incubated with ENTR-601-45. Data are shown as mean ± SD (n=4). Ordinary one-way ANOVA and Dunnett's multiple comparison test; *p<0.05****p<0.0001 relative to untreated DMDΔ46-48.

Exon Skipping with ENTR-601-45 in hDMD Mice

ENTR-601-45 produced improved exon skipping compared to EEV-PMO45 (casimersen[9] PMO sequence conjugated to EEV) in skeletal and cardiac muscle of hDMD mice.

Figure 4. Exon Skipping in hDMD Mice.

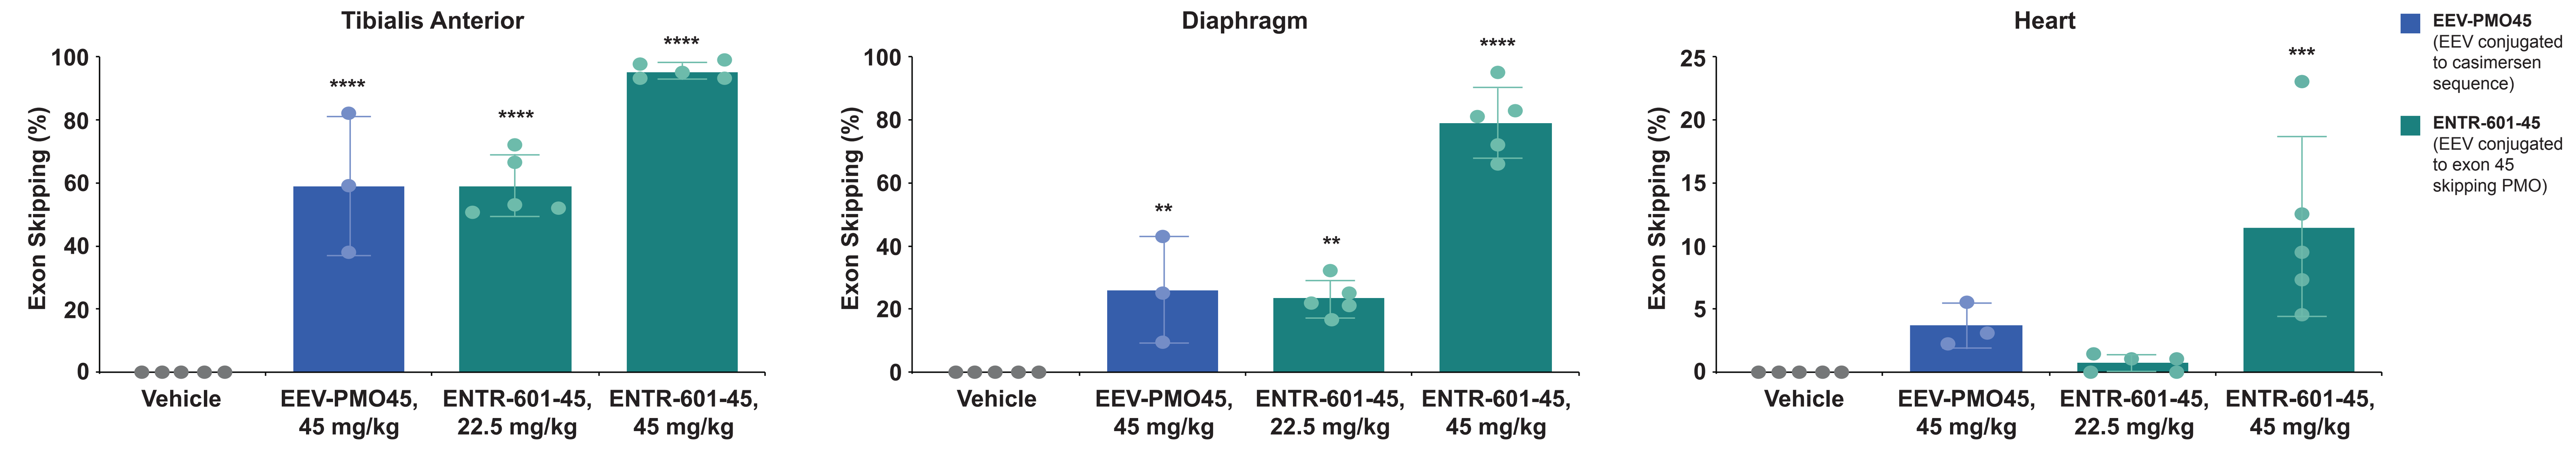
Tibialis Anterior
Diaphragm
Heart
Exon Skipping (%)
Vehicle
EEV-PMO45, 45 mg/kg
ENTR-601-45, 22.5 mg/kg
ENTR-601-45, 45 mg/kg
EEV-PMO45 (EEV conjugated to casimersen sequence)
ENTR-601-45 (EEV conjugated to exon 45 skipping PMO)

hDMD mice were treated with a single intravenous dose of ENTR-601-45 and tissues were harvested 1 week post-injection. Data are shown as mean ± SD (n=3-5); one-way ANOVA; **p<0.01, ***p<0.001, ****p<0.0001; relative to vehicle. Concentrations provided are PMO equivalent. Casimersen is an exon 45 skipping PMO approved in the US.

CONCLUSIONS

- ENTR-601-45 produced robust and dose-dependent exon skipping and dystrophin restoration in patient-derived skeletal and cardiac muscle cells.
- A single dose of ENTR-601-45 resulted in greater exon skipping in skeletal and cardiac muscles of hDMD mice compared with an EEV-casimersen sequence conjugate.
- Together, these findings indicate the potential for further study of ENTR-601-45 in patients with DMD amenable to exon 45 skipping.

ACKNOWLEDGEMENTS

This research was funded by Entrada Therapeutics, Inc (Boston, MA). The authors would like to thank Aji Nair (Entrada) for assistance with poster development and Vlad Batagui (formerly at Entrada) for contributions to this research. Editorial and studio support for this poster was provided by Ashfield MedComms (US), an Inizio company, and was funded by Entrada Therapeutics, Inc. References: 1. Hoffman EP. *Cell.* 1987. 2. Birnkrant DJ. *Lancet Neurol*. 2018. 3. EXONDYS 51® Prescribing Information. 4. VILTEPSO® Prescribing Information. 5. Qian Z. *Biochemistry*. 2016. 6. Sahni A. *ACS Chem Biol*. 2020. 7. Kumar A. MDA 2022. Poster 126. 8. 't Hoen PAC. *J Biol Chem*. 2008. 9. AMONDYS 45® Prescribing Information.

26th Annual Meeting of the American Society of Gene & Cell Therapy (ASGCT)

Entrada Therapeutics, Inc., Boston, MA.